F-759

In Vivo Efficacy of Orally Dosed F901318, in a Murine Model of Disseminated Aspergillosis.

D. LAW[1], J. OLIVER[1], P. WARN[2], A. KENNEDY[1], G. SIBLEY[1], M. BIRCH[1].

[1]F2G Ltd, Lankro Way, Manchester, UK, [2] Euprotec, Manchester Science Park, Manchester, UK.

dlaw@f2g.com
Tel: +44 161 7851284

Abstract

Background: F901318 is the lead candidate of the novel class of orotomide antifungals which act via the inhibition of DHODH, a key enzyme in pyrimidine biosynthesis, a mechanism not shared with current antifungals. F901318 has a mould only spectrum and has excellent *in vitro* activity against all the clinically significant *Aspergillus* spp. Pharmacokinetic testing shows excellent oral bioavailability in several laboratory animal species. Here we examine the efficacy of orally dosed F901318 in a murine model of disseminated aspergillosis.
Methods: A temporary neutropenic CD-1 mouse model of disseminated aspergillosis was used. Cyclophosphamide treated mice were infected IV with a lethal challenge of *A. fumigatus* A1163 condidia. F901318 treatment commenced 24 h post infection and animals were treated for 8 days and observed for 1 days following cessation of treatment. F901318 was dosed orally as a solution in PEG300 twice daily. Blood samples were taken for galactomannan index (GI) measurement from mice succumbing to infection during the study and from surviving mice at the end of the study.
Results: Untreated animals all died of the infection by day 6 whereas there was a clear dose dependent response observed for F901318. A dose of 1mg/kg BD showed no effect on survival, 3mg/kg BD showed 60% survival and 10mg/kg BD gave 100% survival. There was also a dose dependent response in GI, a high GI was seen in untreated animals and mice dosed with 1mg/kg BD F901318, lower GI values were seen in mice treated with 3mg/kg BD and very low GI was seen in mice treated with 10mg/kg BD F901318.
Conclusions: F901318 dosed orally to mice with disseminated aspergillosis demonstrated a dose dependent response in survival. A dose of 10mg/kg BD conferred complete survival in this model and was also effective in significantly reducing galactomannan levels in infected mice. Reduction in galactomannan indices are known to correlate with a beneficial outcome in human invasive aspergillosis.
F901318 has demonstrated excellent antifungal activity when dosed orally in a disseminated mouse model of aspergillosis and further development of this compound is warranted.

Introduction

F901318 is the lead candidate of the novel class of orotomide antifungals which act via the inhibition of DHODH, a key enzyme in pyrimidine biosynthesis. F901318 has excellent *in vitro* activity against all the clinically significant *Aspergillus* spp. Pharmacokinetic studies show that F901318 has excellent oral bioavailability in several laboratory animal species. In this study we examine the efficacy of orally dosed F901318 in a murine model of disseminated aspergillosis.

Methods

Groups of 10 CD-1 mice were immunosuppressed with cyclophosphamide (200mg/kg), 3 days later mice were infected through the tail vein with a lethal challenge of *A. fumigatus* A1163 conidia. Treatment with F901318 commenced 24hr post infection and continued for 8 days, animals were observed for a further day following cessation of treatment. F901318 was dosed orally in a solution of PEG300. Three different doses of F901318 were evaluated, 1mg/kg, 3mg/kg or 10mg/kg each dosed PO twice daily. Survival was monitored on a daily basis. Blood samples were taken from mice succumbing to infection during the study or from surviving mice at the end of the study. Galactomannan (GM) was measured in serum using the Platelia™ Aspergillus Ag kit (Bio-Rad). Samples were diluted 1 in 5 with water prior to testing as preliminary data indicated very high levels in untreated animals.

Fig. 1 The structure of F901318

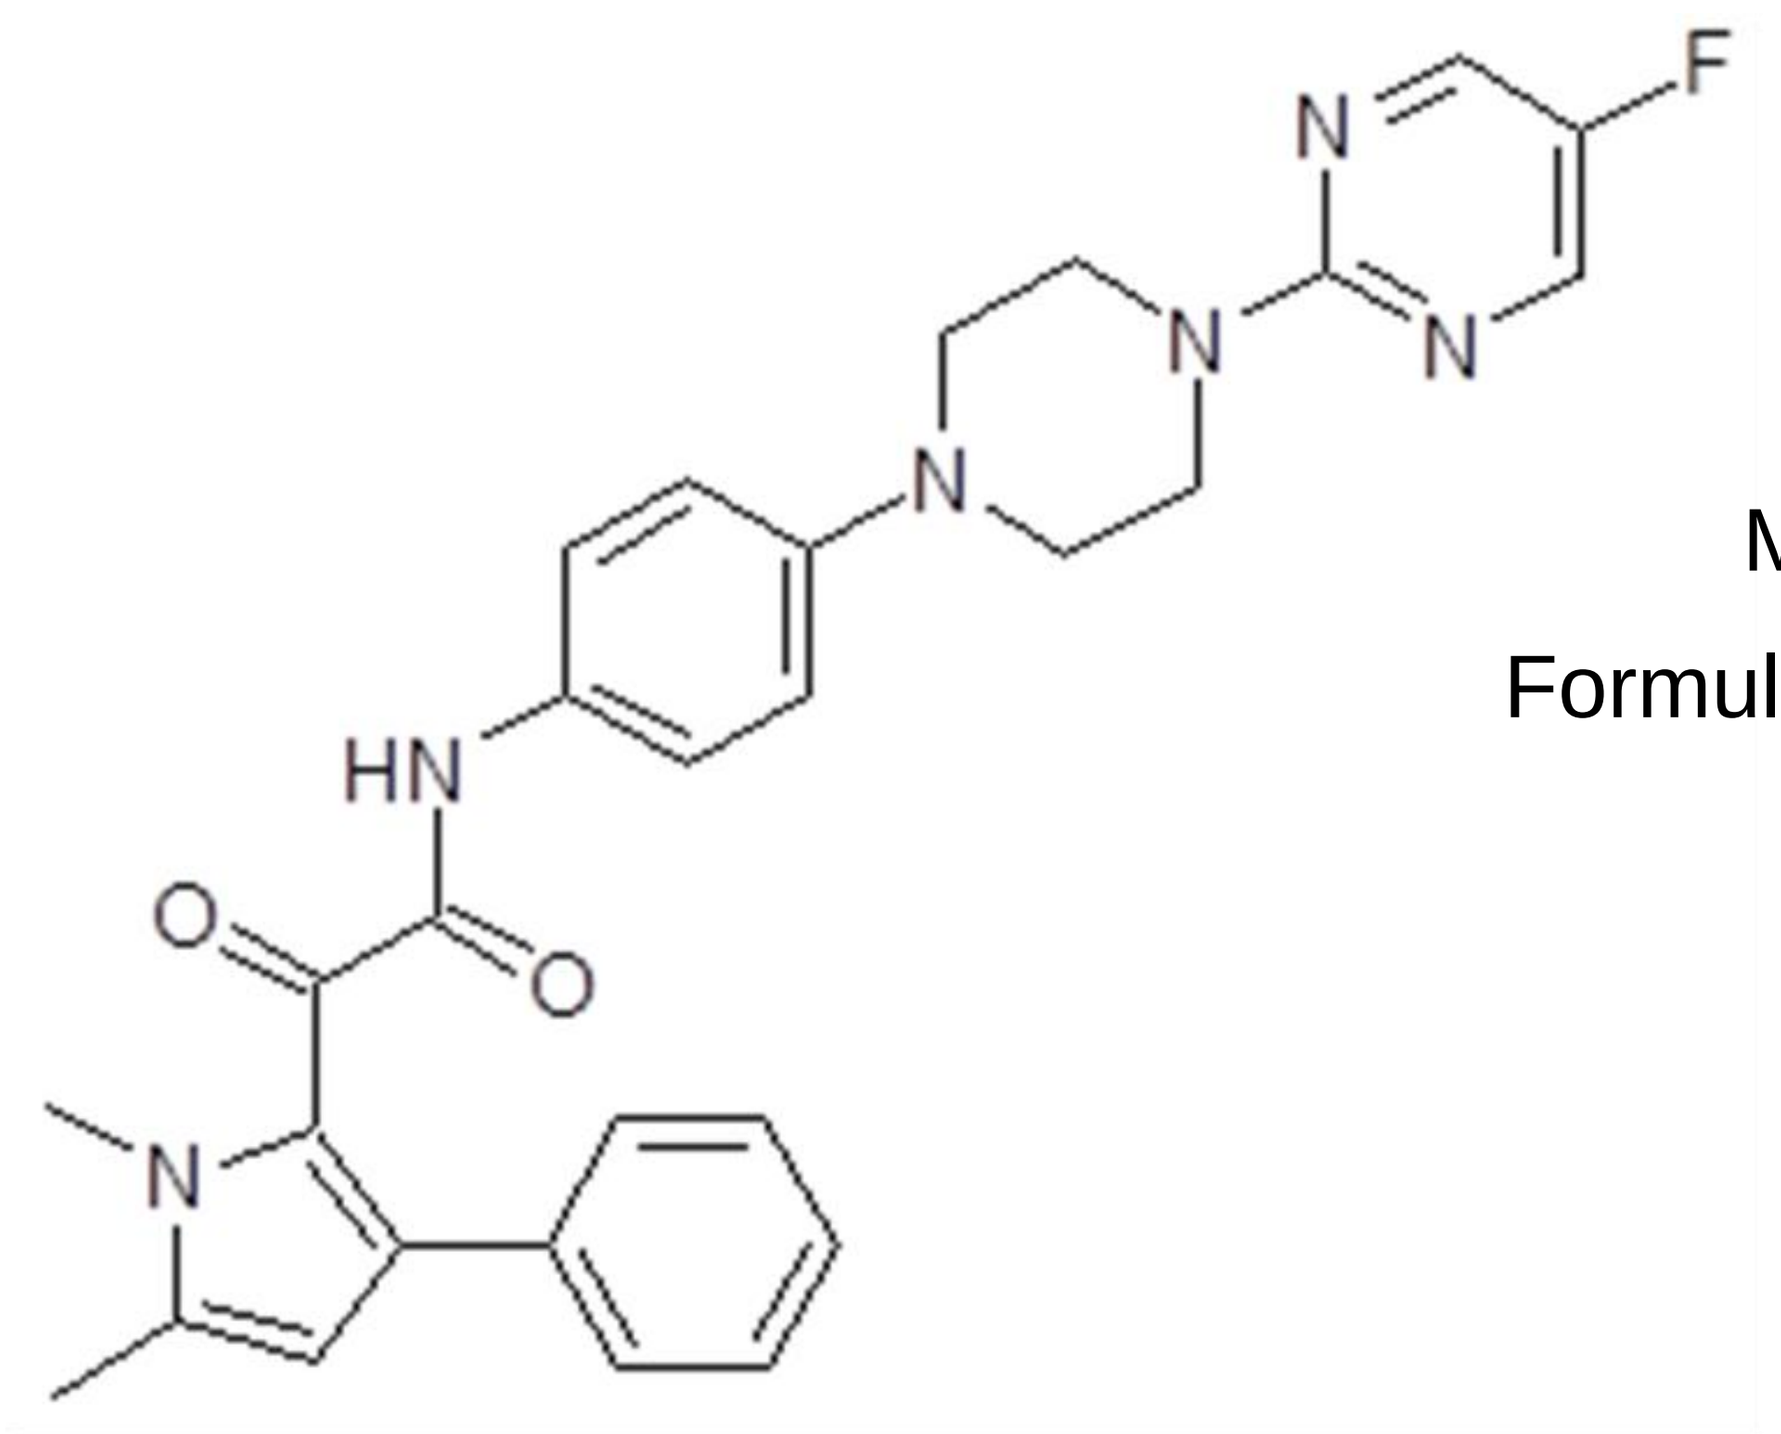

N
F
N
N
N
HN
O
O
N

MW = 499

Formula = $C_{28}H_{27}FN_6O_2$

Results

Fig. 2 Survival graph for treatment groups

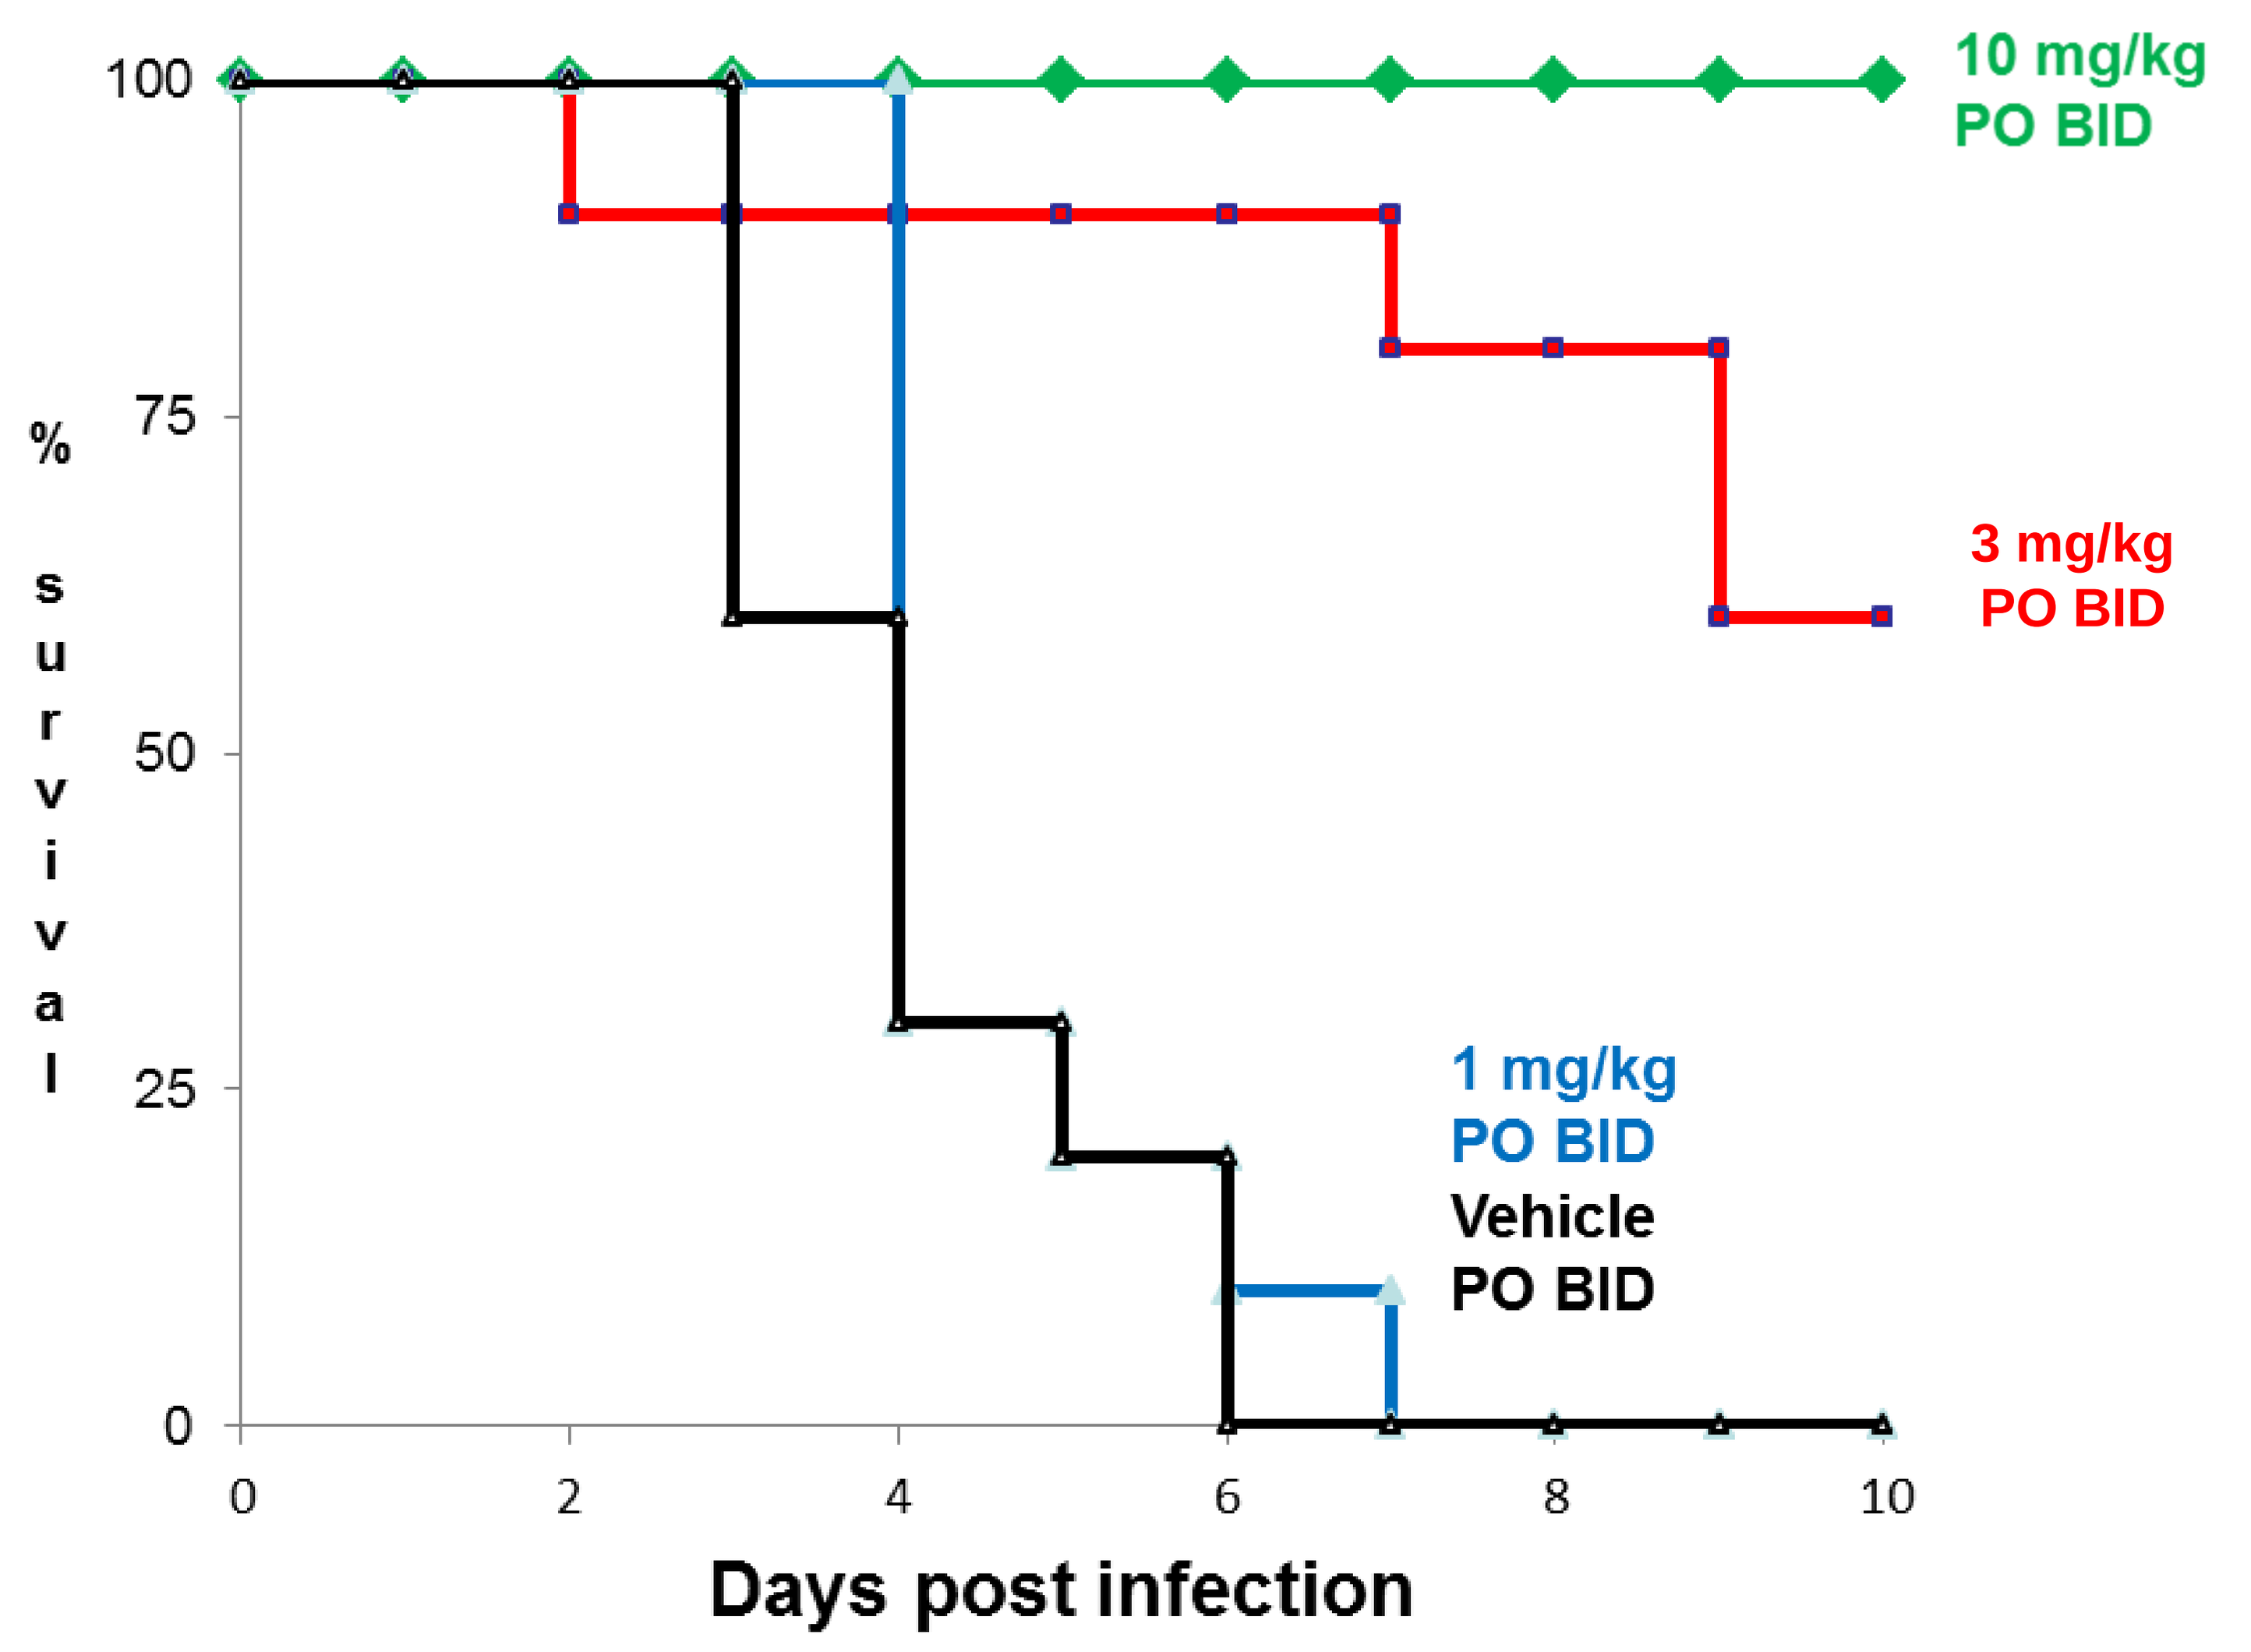

100
75
50
25
0
% survival
0
2
4
6
8
10
Days post infection
10 mg/kg
PO BID
3 mg/kg
PO BID
1 mg/kg
PO BID
Vehicle
PO BID

Data in Figure 2 shows survival for each treatment group. Whilst all untreated animals succumbed to infection by day 6 there is a clear dose response for F901318. A dose of 1mg/kg BID had no effect on survival whilst a dose of 10mg/kg BID gave 100% survival at the end of the study. The intermediate dose of 3mg/kg BID gave 60% survival at end of study.

Figure 3 shows the mean galactomannan (GM) index in the various treatment groups either at end of study or at time of death if earlier. (Error bars indicate standard deviation).

Fig. 3 Mean galactomannan index from each treatment group

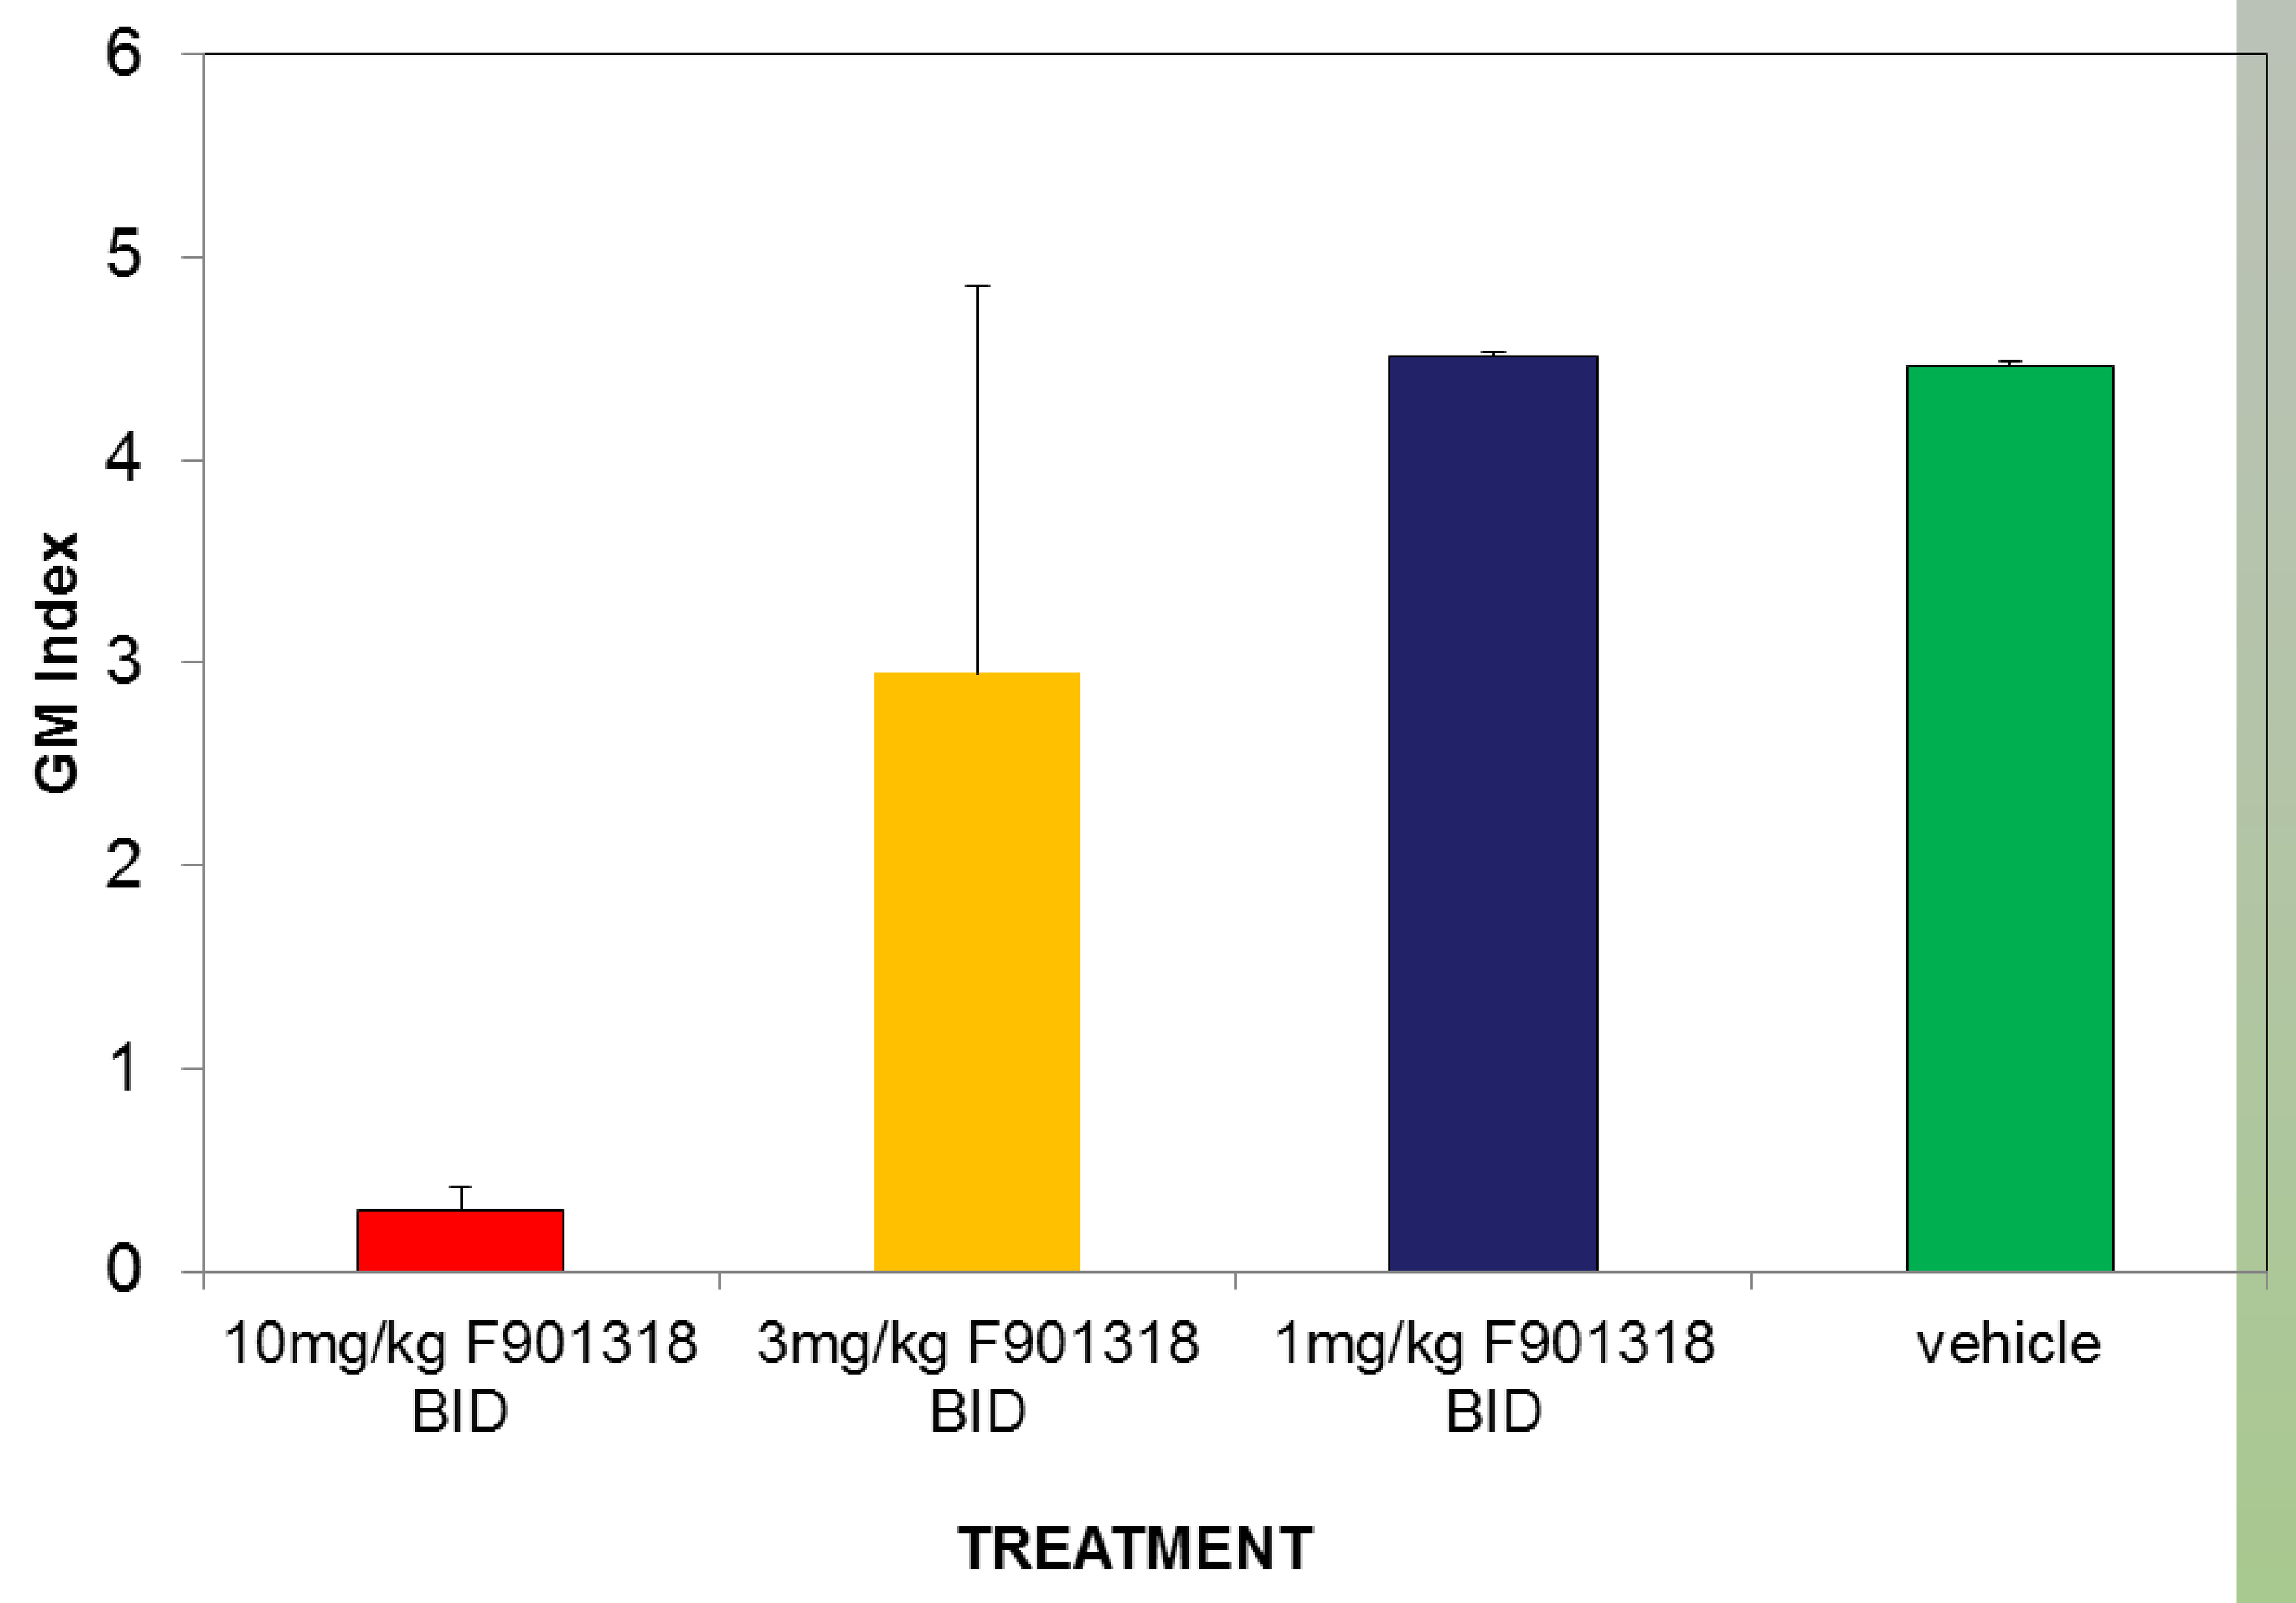

6
5
4
3
2
1
0
GM Index
10mg/kg F901318 BID
3mg/kg F901318 BID
1mg/kg F901318 BID
vehicle
TREATMENT

There is a clear dose dependent response in the mean GM index for each group which correlates well with survival data. Very low GM indices were observed in mice treated with 10mg/kg BID F901318, which survived to the end of the study. Mice treated with 1mg/kg BID F901318 had high GM indices similar to those in untreated animals. The 3mg/kg BID treatment group had an intermediate mean GM index reflecting the survival data.

Conclusions

In a severe model of disseminated invasive aspergillosis, orally administered F901318 shows a clear dose response in terms of improving survival and reducing the serum galactomannan index.

An oral dose of 10mg/kg BID gave a survival rate of 100% at the end of study, and produced a marked reduction in galactomannan index.

As the GM index is a useful biomarker for monitoring therapy of aspergillosis in humans, the ability of F901318 to reduce the GM index in a model of murine invasive aspergillosis suggests that measurement of the GM index could be used to monitor the efficacy of F901318 in humans.

Printed by Call4Posters