F-756

Spectrum of Activity of F901318, the First Agent from the New Orotomide Class of Antifungals

A. FOTHERGILL[1], N. WIEDERHOLD[1], G. SIBLEY[2], A. KENNEDY[2], J. OLIVER[2], M. BIRCH[2], D. LAW[2].

[1] University of Texas at San Antonio, TX., USA; [2] F2G Ltd, Manchester, UK.

fothergill@uthscsa.edu
Tel: 210 567 4131.

Abstract

Background: F901318 is the most advanced analog of the orotomide class of antifungal agents and acts via a novel mode of action. F901318 is in clinical development for the treatment of serious systemic fungal infections, in particular invasive aspergillosis. In this study the antifungal spectrum of F901318 was assessed by carrying out susceptibility testing against a range of pathogenic yeasts and moulds to determine the spectrum of activity **Methods**: The testing methods used in this study are as outlined in CLSI documents M38-A2 and M27-A3. All organisms were tested in a broth dilution format (concentration range 32-0.06 mg/L). **Results:** No activity was detected (MIC >32mg/L) against *Candida albicans*, *C. krusei*, C. *glabrata*, *C. tropicalis,* C. *parapsilosis* (n=4 all species) or *Cryptococcus neoformans* (n=2). However, F901318 had potent activity against the common species of *Aspergillus, A. fumigatus, A. flavus, A. terreus* and *A. niger* (n=25 for all species) with Geomean MICs of 0.05, 0.04, 0.013 and 0.08 mg/L respectively. In addition, potent activity (MICs <0.06 mg/L) was also observed against *Aspergillus lentulus* (n=4), a cryptic species often associated with increased resistance to azoles. F901318 also demonstrated potent activity (MICs <0.06 mg/L) against *Lomentospora (Scedosporium) prolificans* (n=3), *Scedosporium apiospermum* (n=4) in addition to several hyaline fungi, including *Paecilomyces* spp., *Acremonium* spp., *Scopulariopsis* spp. and *Penicillium marneffei* (n= 5 for each). Activity was also noted against some *Fusarium* spp. isolates (n=5) although MICs were higher (0.25-4mg/L). When tested against the agents of the endemic mycoses (n=5 for each species), F901318 MICs were <0.06 mg/L against *Blastomyces dermatitidis* and *Coccidiodes immitis/ posadasii.* For *Histoplasma capsulatum,* MICs were 0.06-0.125 mg/L. No activity was observed against the Mucorales or *Alternaria alternata* (MICs >32 mg/L). **Conclusion**: In this study, F901318 was shown to lack activity against *Candida* spp. but demonstrated potent activity against a spectrum of filamentous fungi including many pathogenic species that are resistant to current antifungal agents. These include *L. prolificans* and *Fusarium solani* for which effective therapies are lacking.

Introduction

Invasive fungal infections remain a significant clinical problem in high-risk populations, including heavily immunocompromised patients, those with multiple comorbidities, and patients in intensive care settings. Thus, there is a need for the development of new antifungal agents.

F901318 is the most advanced analog of the orotomide class of antifungal agents. This investigational agent acts via a novel mode of action namely the selective inhibition of DHODH a critical enzyme in pyrimidine biosynthesis.

The objective of this study was to determine the *in vitro* spectrum of activity of F901318 against different pathogenic fungi known to cause disease in humans. This included yeasts, moulds, and endemic fungi.

Methods

Clinical yeast and mould isolates were obtained from the Fungus Testing Laboratory at the UT Health Science Center at San Antonio.

In vitro antifungal activity was measured according to the CLSI M27-A3 and M38-A2 guidelines (1, 2). Broth microdilution susceptibility testing was performed for all species, except *Blastomyces dermatitidis*, *Coccidioides* spp., and *Histoplasma capsulatum*, for which macrodilution susceptibility testing was performed.

Against *Aspergillus flavus, A. fumigatus, A. niger,* and *A. terreus*, susceptibility testing was also performed with amphotericin B (AMB), caspofungin (CAS), and voriconazole (VOR). The MICs for F901318, amphotericin B, and voriconazole were read as the lowest concentration that inhibited 100% of growth. For caspofungin, the minimum effective concentration (MEC) was the lowest concentration that resulted in morphologic abnormalities.

Against *Aspergillus* spp., the MICs that inhibited 50% and 90% of the fungi (MIC50 and MIC90, respectively), and the geometric mean (GM) MICs were determined. Differences in GM MIC values were assessed for significance by ANOVA with Tukey's post-test for multiple comparisons.

Results

The MIC distributions for each agent against all *Aspergillus* isolates are depicted in **Figure 1**. F901318 demonstrated potent *in vitro* activity against all *Aspergillus* species. The GM MIC of F901318 at 100% inhibition of growth after 48 hours was compared to those of the other agents against all isolates (**Table 1)**. In this comparison, the GM MIC of F901318 was significantly lower than either amphotericin B and voriconazole.

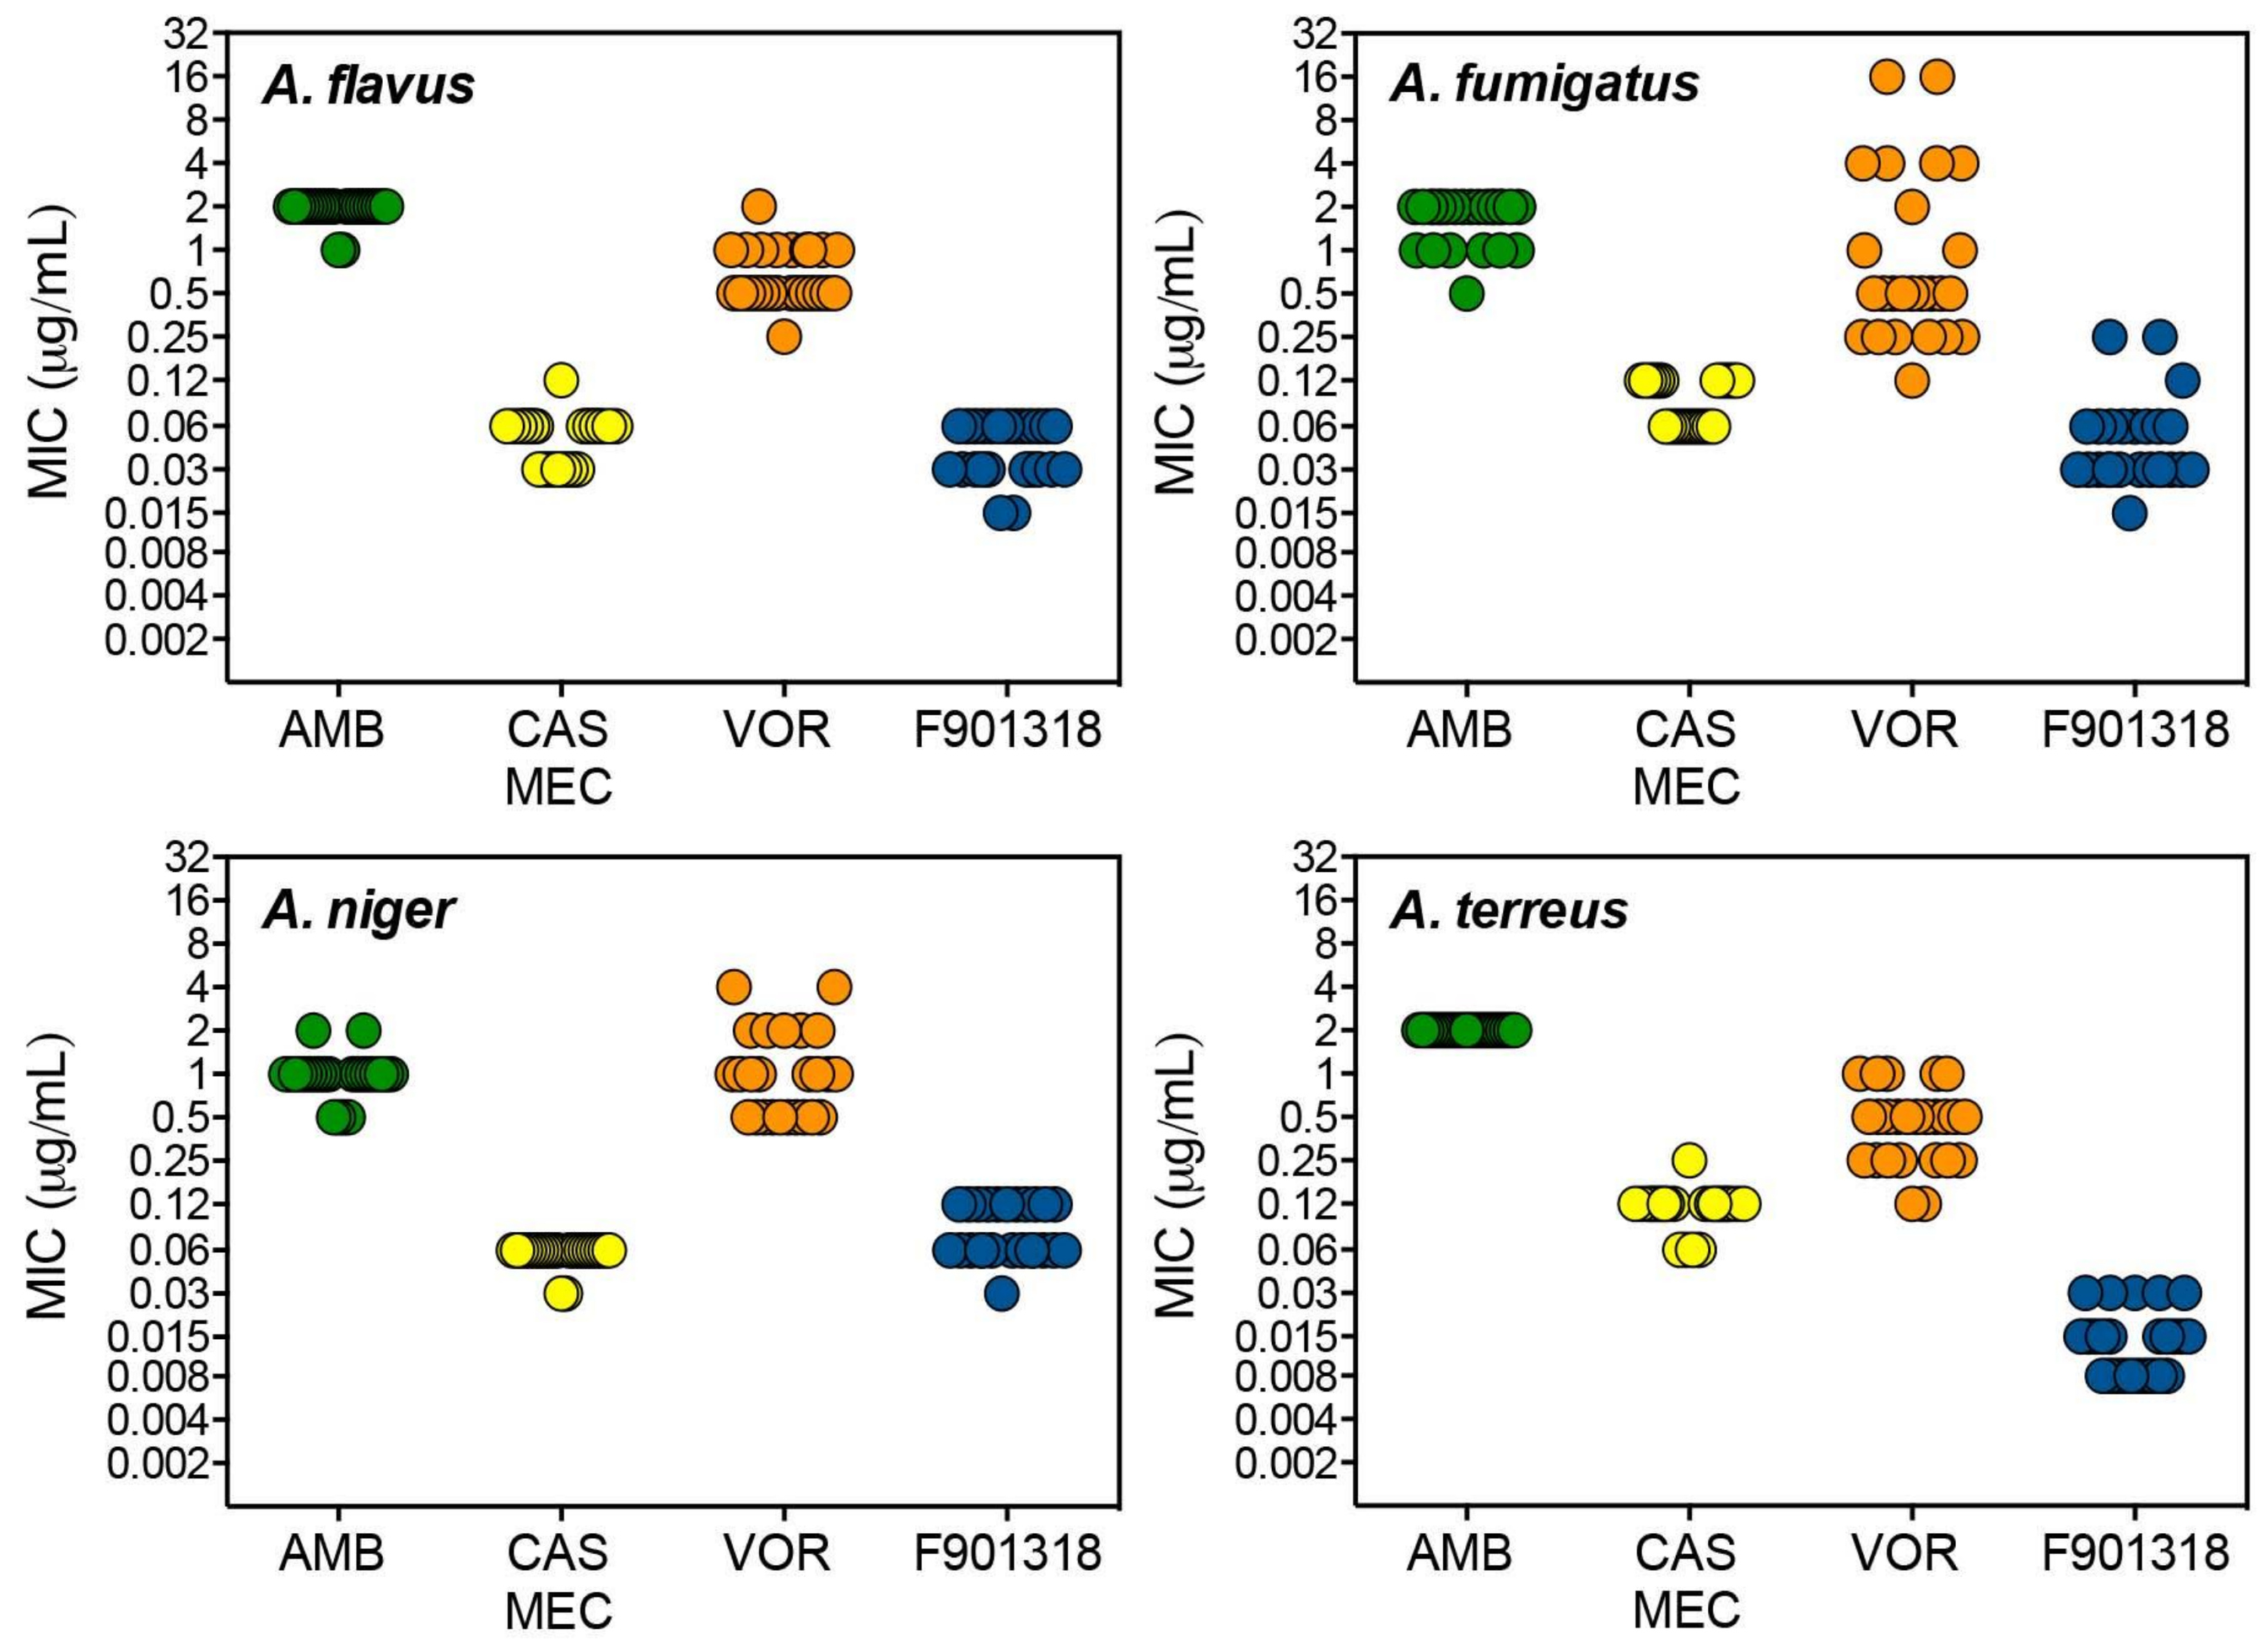
A. flavus
A. fumigatus
A. niger
A. terreus
MIC (µg/mL)
32
16
8
4
2
1
0.5
0.25
0.12
0.06
0.03
0.015
0.008
0.004
0.002
AMB
CAS MEC
VOR
F901318

Figure 1. MIC/MEC distributions of amphotericin B, (AMB) caspofungin (CAS), voriconazole (VOR), and F901318 against *Aspergillus* species, including *A. flavus*, *A. fumigatus*, *A. niger*, and *A. terreus* (n = 25 isolates per species).

A. flavus (n = 25)

Agent	AMB	CAS (MEC)	VOR	F901318
MIC Range	1 - 2	0.03 - 0.12	0.25 - 2	0.015-0.06
MIC50	2	0.06	0.5	0.06
MIC90	2	0.06	1	0.06
GM MIC	1.548 (p<0.0001)	0.0728	0.6878 (p<0.0001)	0.0376

A. fumigatus (n = 25)

Agent	AMB	CAS (MEC)	VOR	F901318
MIC Range	0.5 - 2	0.06 - 0.12	0.12 - 16	0.015-0.25
MIC50	2	0.06	0.5	0.03
MIC90	2	0.125	16	0.25
GM MIC	1.602 (p<0.0001)	0.0853	0.8236 (p<0.0001)	0.0457

A. niger (n = 25)

Agent	AMB	CAS (MEC)	VOR	F901318
MIC Range	0.5 - 2	0.03 - 0.06	0.5 - 1	0.03-0.12
MIC50	1	0.06	1	0.06
MIC90	2	0.06	4	0.125
GM MIC	0.9461 (p<0.0001)	0.0568	0.9727 (p<0.0001)	0.0806

A. terreus (n = 25)

Agent	AMB	CAS (MEC)	VOR	F901318
MIC Range	2	0.06 - 0.12	0.12 - 1	0.008-0.03
MIC50	2	0.125	0.5	0.015
MIC90	2	0.125	1	0.03
GM MIC	2.000 (p<0.0001)	0.1143	0.4234 (p<0.0001)	0.0134

p-values vs. F901318 GM MIC

Table 1. MIC/MEC distributions, MIC50 & 90 values, and GM MIC values of amphotericin B (AMB), caspofungin (CAS), voriconazole (VOR), and F901318 against *A. flavus*, *A. fumigatus*, *A. niger*, and *A. terreus.*

F901318 also demonstrated potent activity against *A. lentulus,* a cryptic species often associated with increased resistance to azoles. Potent *in vitro* activity was also observed against other moulds, including species that are known or have been shown to have decreased susceptibility or are frequently resistant to other antifungals, including *Lomentospora (Scedosporium) prolificans*, *Scopulariopsis,* and *Fusarium* species (**Figure 2)**. However, no activity was observed against *Candida* spp., *Cryptococcus neoformans*, members of the Order *Mucorales* or *Alternaria alternata* (MIC > 32 mg/L)

Figure 2. MIC distributions for F901318 against other mould species

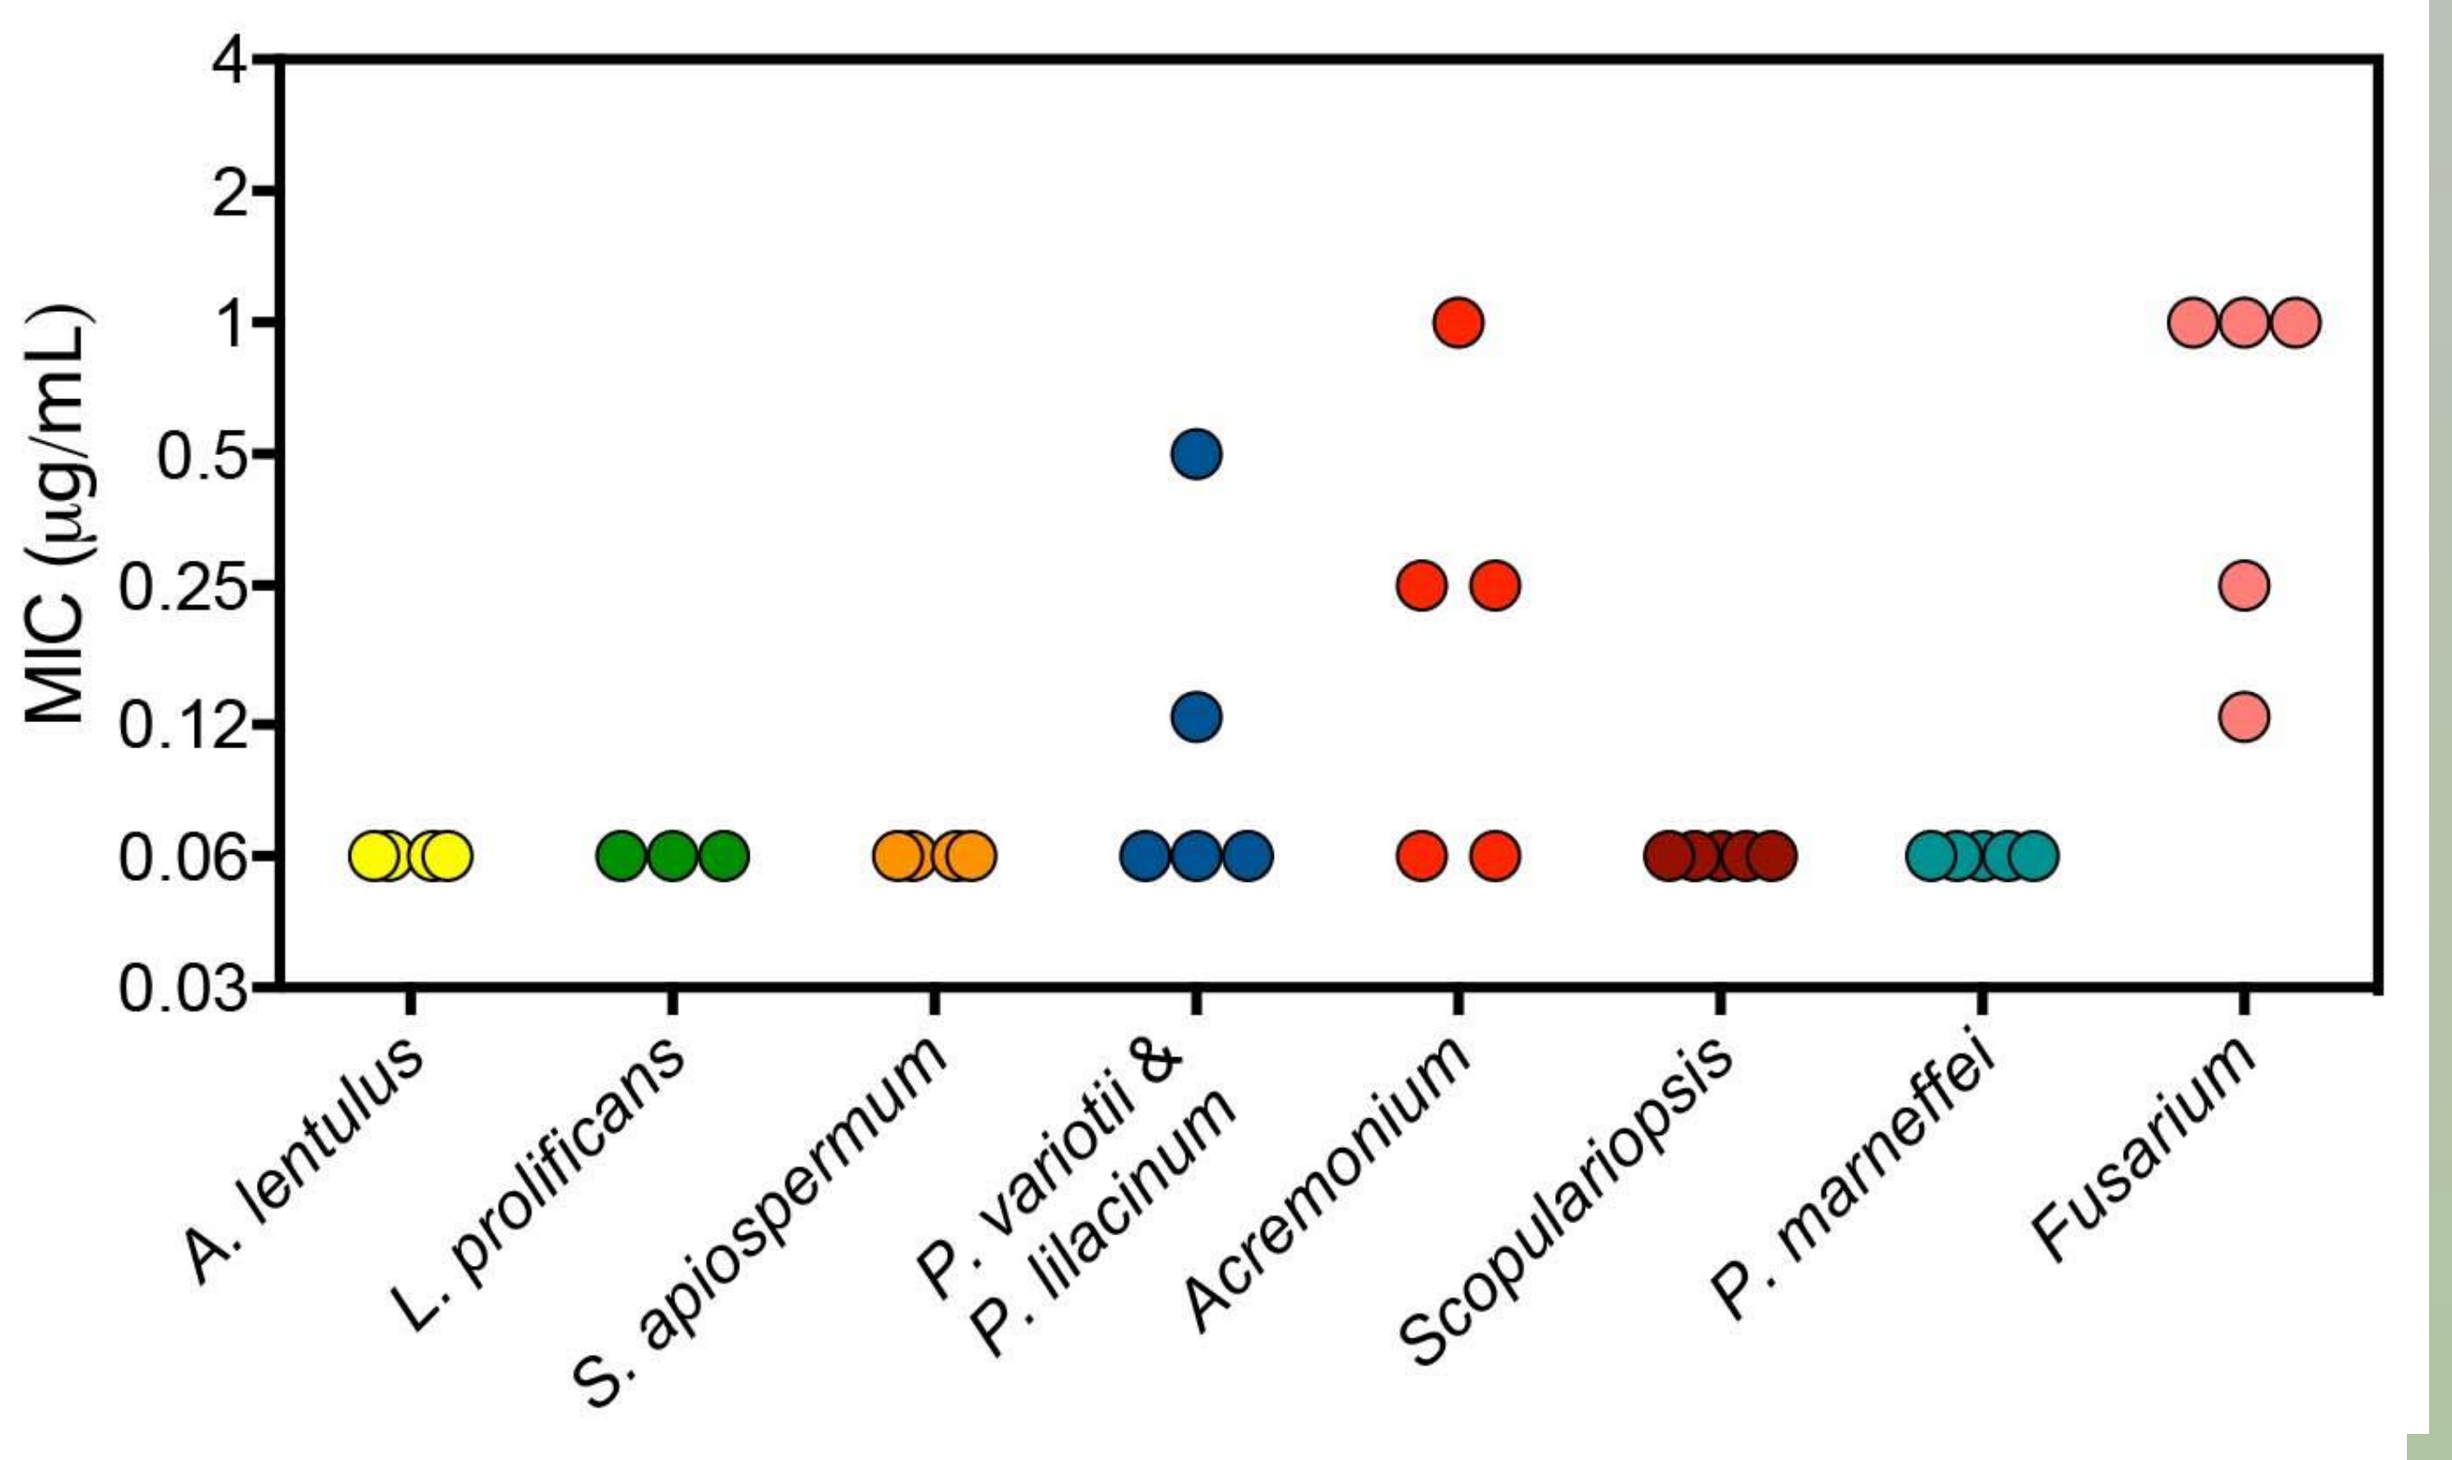
MIC (µg/mL)
4
2
1
0.5
0.25
0.12
0.06
0.03
A. lentulus
L. prolificans
S. apiospermum
P. variotii & P. lilacinum
Acremonium
Scopulariopsis
P. marneffei
Fusarium

F901318 also demonstrated potent activity against endemic species, including *Blastomyces dermatitidis, Coccidioides immitis/posadasii*, and *Histoplasma capsulatum* (**Figure 3)**.

Figure 3. MIC distributions for F901318 against endemic fungi.

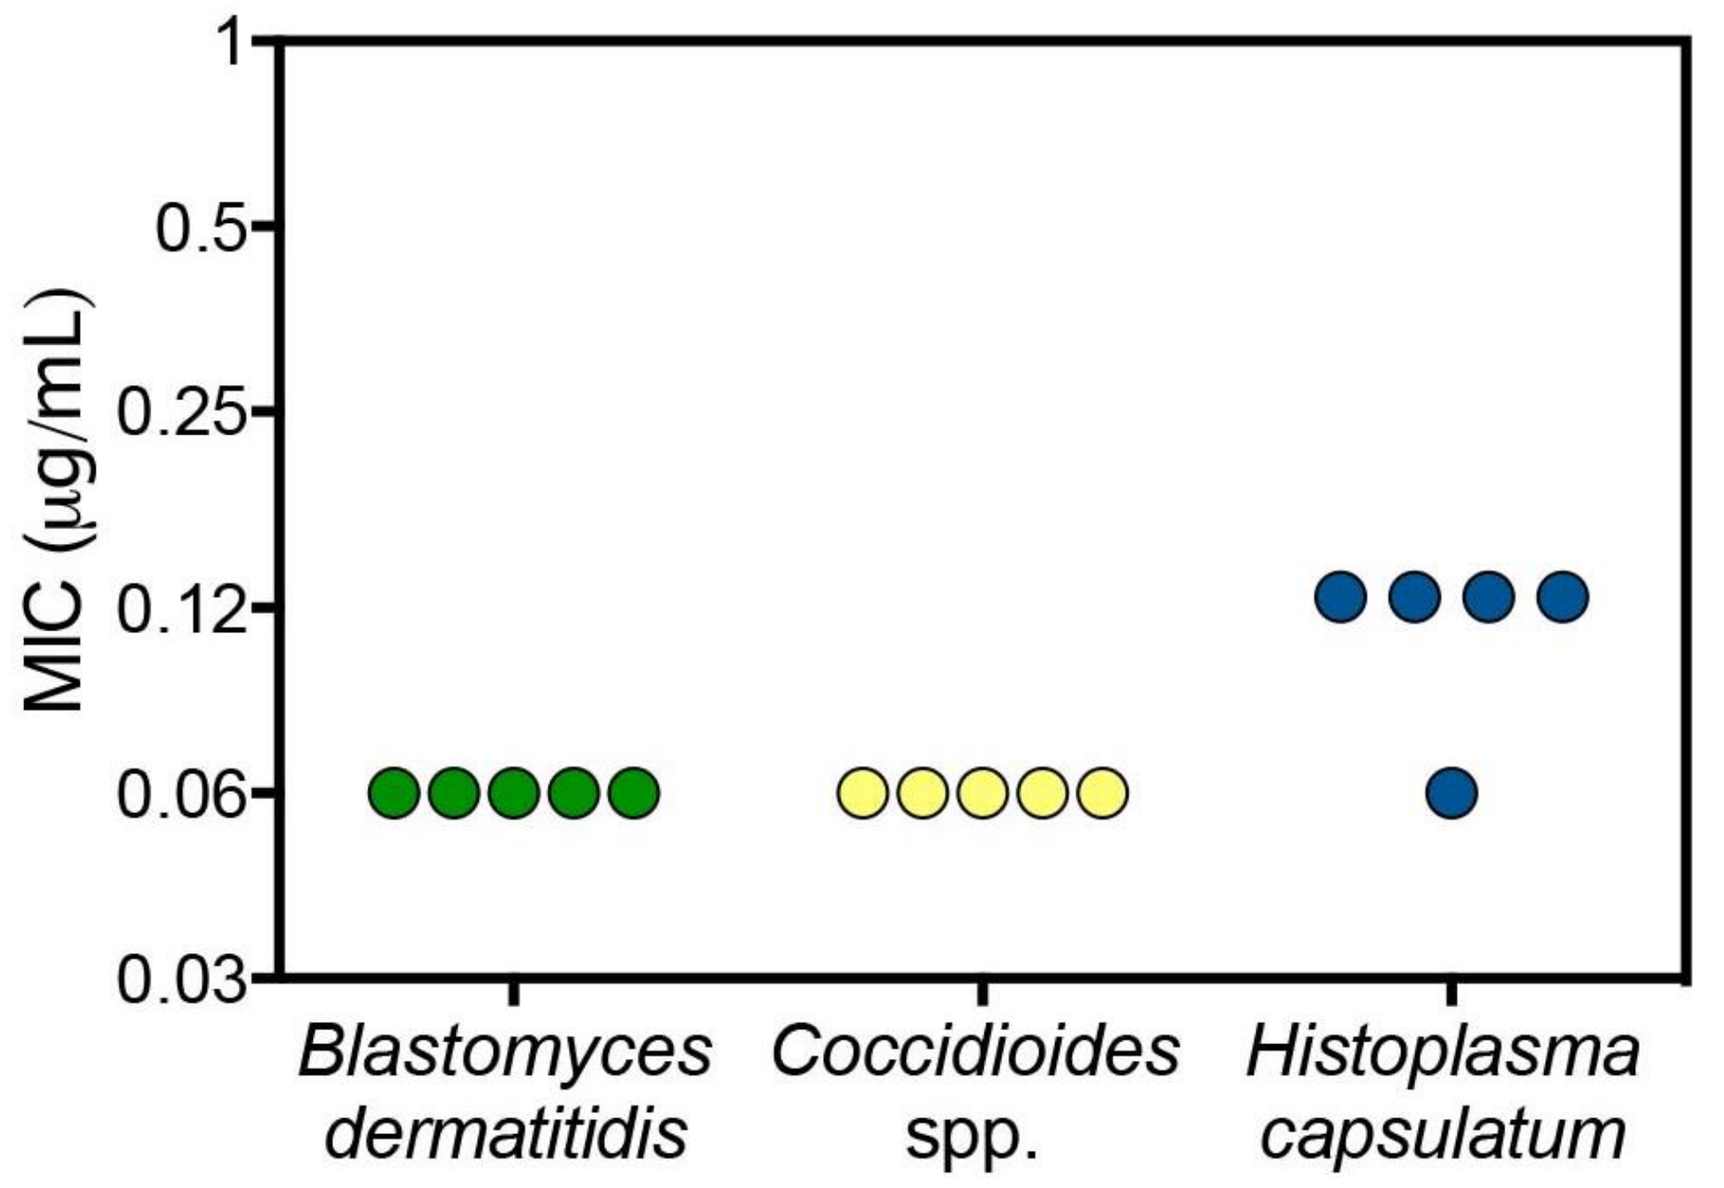
MIC (µg/mL)
1
0.5
0.25
0.12
0.06
0.03
Blastomyces dermatitidis
Coccidioides spp.
Histoplasma capsulatum

Conclusions

F901318 demonstrated potent *in vitro* activity against various pathogenic moulds and endemic fungi. The activity of this agent was also maintained against several species that are usually resistant to other antifungals, including *A. lentulus, L. (Scedosporium) prolificans, S. apiospermum, Scopulariopsis* spp., and *Fusarium* spp. F901318 was also significantly more potent than amphotericin B and voriconazole against *A. flavus, A. fumigatus, A. niger,* and *A. terreus.*

References

1) Clinical Laboratory Standards Institute, 2008. Reference method for broth dilution antifungal susceptibility testing of filamentous fungi; approved standard, 2nd ed. CLSI document M38-A2. Clinical and Laboratory Standards Institute, Wayne, PA.

2) Clinical and Laboratory Standards Institute. 2008. Reference method for broth dilution antifungal susceptibility testing of yeasts, 3rd ed. CLSI document M27-A3. Clinical and Laboratory Standards Institute, Wayne, PA.

Funding and F901318 powder was provided by F2G, Ltd

Printed by Call4Posters