#P1704

ECCMID 2017

IN VITRO SUSCEPTIBILITY TESTING OF THE NOVEL OROTOMIDE ANTIFUNGAL AGENT F901318 AGAINST AUSTRALIAN *SCEDOSPORIUM* AND *LOMENTOSPORA* PATHOGENS.

C. Biswas[1], S. Chen[1], C. Halliday[1], M. Slavin[2], M. Birch[3], D. Law[3] [1]Westmead Hospital, Sydney, Australia, [2]Peter MacCallum Cancer Centre, Melbourne, Australia. [3]F2G Ltd, Manchester, UK.

Abstract

Background: Infections caused by organisms *Scedosporium* spp. and *Lomentospora* (formerly *Scedosporium*) *prolificans* are the second commonest non-*Aspergillus* mould infections in Australia. Treatment is problematic because these fungi, in particular *L. prolificans,* are intrinsically resistant to currently available antifungals. New antifungal agents are urgently required. F901318 is an antifungal agent with a novel mechanism of action currently completing Phase I clinical development. The objective of this study was to compare the *in vitro* potency of F901318 against Australian clinical isolates of *Scedosporium* and *L. prolificans* with standard antifungals.
Material/methods: Fifty clinical *Scedosporium* isolates (10 *S. apiospermum,* 7 *S. aurantiacum,* 3 *S. boydii* and 30 *L. prolificans)* were studied. Each isolate was identified by morphologic assessment and sequencing of the ITS gene region. Susceptibility testing of F901318 and isavuconazole was performed according to the CLSI M38-A2 reference standard. Susceptibility to other agents (amphotericin B, posaconazole, voriconazole, itraconazole, fluconazole, micafungin, caspofungin and anidulafungin) were performed using Sensititre plates. Four *Aspergillus fumigatus* isolates were included as comparators.
Results: The table shows MIC data for some of the drugs tested in this study. F901318 demonstrated the most potent *in vitro* activity of the agents tested. F901318 was active against all isolates of *L. prolificans* strains with MICs falling into a very narrow range (0.125-0.5mg/L). In contrast, the *L. prolificans* isolates showed a high level of resistance to the other agents tested. F901318 MICs against the three species of *Scedosporium* also fell into a narrow range (0.125-0.5mg/L). The activity of standard antifungals was variable against these three species with only voriconazole and itraconazole active against all of the strains. F901318 MICs for *Scedosporium* and *Lomentospora* spp. were very similar to those obtained for 4 *A. fumigatus* isolates.
Conclusions: In this study, F901318 demonstrated the most potent *in vitro* activity against 50 strains representing three different species of *Scedosporium* inclusive of 30 strains of *L. prolificans*, and was the only agent to show consistently low MICs against the latter. This data confirms activity seen in previous studies but extends the findings to a larger panel of *L. prolificans* isolates from a different geographical location supporting generalisability of data.

Introduction

Invasive fungal disease due to uncommon mould pathogens are on the rise. Of these, *Scedosporium* species and *Lomentospora* (previously *Scedosporium*) *prolificans* account for a significant proportion of cases. In Australia, they are the second most common cause (30% of cases) of non-*Aspergillus* mould infections after the Mucorales. Because these fungi are frequently resistant to almost all available antifungals, new agents are urgently needed.
The novel orotomide agent, F901318, has shown promising *in vitro* and *in vivo* results in early stage testing against a small number of *Scedosporium/Lomentospora* species.

Objective
The study objectives were to determine the *in vitro* activities of F901318 against *Scedosporium* and *Lomentospora* fungi in comparison with standard antifungal drugs used to treat invasive mould infections, including the newest azole, isavuconazole.

Methods

Fungal isolates
Fifty clinical isolates of *Scedosporium*/*Lomentospora* species were studied. These comprised 30 *L. prolificans*, and 20 members of the *S. apiospermum* species complex (10 *S. apiospermum sensu stricto*, 3 *S. boydii*, and 7 *S. aurantiacum*). The species identity of all the strains was confirmed by sequencing of the fungal ITS rRNA gene region. In addition, four *Aspergillus fumigatus* strains were also tested including *A. fumigatus* ATCC 20435.

Drugs
F901318, amphotericin B (**AMB**), caspofungin (**CAS**), anidulafungin (**ANI**), micafungin (**MIC**), itraconazole, (**ITC**), voriconazole (**VRC**), posaconazole (**POS**), isavuconazole (**ISV**),. fluconazole (**FLU**) and 5-flucytosine (**5FC).**

Susceptibility testing
Drug dilutions used were: **AMB**, 0.12 to 8 mg/L; **5FC**, 0.06-64 mg/L; **FLU**, 0.12-256 mg/L; **ITC**, 0.015-16 mg/L; **VRC**, **POS**, **CAS**, **ANI**, and **MIC** 0.008-8 mg/L, **ISV** 0.06-32 mg/L and **F901318** 0.004 -2 mg/L.

Susceptibility testing of F901318 and isavuconazole against the study isolates was performed according to CLSI M38-A2 reference methodology for susceptibility testing of filamentous fungi. For testing of other agents, the Sensititre Yeast Y010 method was used. Minimum inhibitory concentrations (MICs) and minimum effective concentrations (MECs) in the case of the echinocandins were determined according to the CLSI criteria for these drugs, where available. For F901318, the MIC was taken as the minimum drug concentration required to inhibit growth of the organism at 100%.

Each isolate was tested in duplicate on two separate occasions. Each run included standard CLSI quality control strains.

Results

The MIC ranges and geometric mean MICs for each species are shown in the table. The MIC distributions for all isolates are shown in figures 1, 2 and 3.

F901318 was the most potent of the drugs tested and low MICs were recorded against all isolates including *L. prolificans*. with MICs against this species falling into a very narrow range (0.125-0.5mg/L). For comparison F901318 MICs against 4 isolates of *A. fumigatus* were 0.125mg/L.

MICs to the 3 echinocandin drugs were similar for all 3 drugs and were higher than those of F901318. Amphotericin B MICs were high for all four species. For fluconazole and flucytosine the majority of *Scedosporium* and *Lomentospora* strains had very high MICs (16 - >256 mg/L) as expected.

MICs against the azole drugs were variable - most isolates of *L. prolificans* had high MICs against all of the azoles. Itraconazole, voriconazole and posaconazole had lower MICs against the three *Scedosporium* species tested and MICs were in a similar range to those of F901318. However, isavuconazole was much less active than the other azoles against all 4 test species.

Only F901318 had consistently low MICs against all 30 isolates of *L. prolificans.*

Table

Fungus	n		F901318	AMB	CAS	ANI	MIC	ITC	VRC	POS	ISV	FLU	5FC
Scedosporium apiospermum	10	GM	0.15	3.06	1.44	6.94	10.56	0.34	0.12	0.55	5.28	12.13	78.79
		Range	0.125-0.5	2-16	0.12-16	2-16	4-16	0.25-1	0.06-0.5	0.25-128	2-8	2-256	1-128
Scedosporium aurantiacum	7	GM	0.25	9.75	4.42	5.94	5.38	0.61	0.11	0.37	8	8.83	128
		Range	0.125-0.5	2-16	2-16	4-16	0.5-16	0.12-32	0.03-0.25	0.12-1	4-16	2-512	128
Scedosporium boydii	3	GM	0.2	6.35	8	16	16	0.5	0.12	0.5	5.04	16	128
		Range	0.125-0.25	4-8	8	16	16	0.5	0.12	0.5	4-8	16	128
Lomentospora prolificans	30	GM	0.26	11.31	7.64	7.29	8.39	32	5.66	16	16.76	512	128
		Range	0.125-0.5	4-16	4-16	2-16	2-16	32	0.5-16	16	8-32	512	128

Figure 1

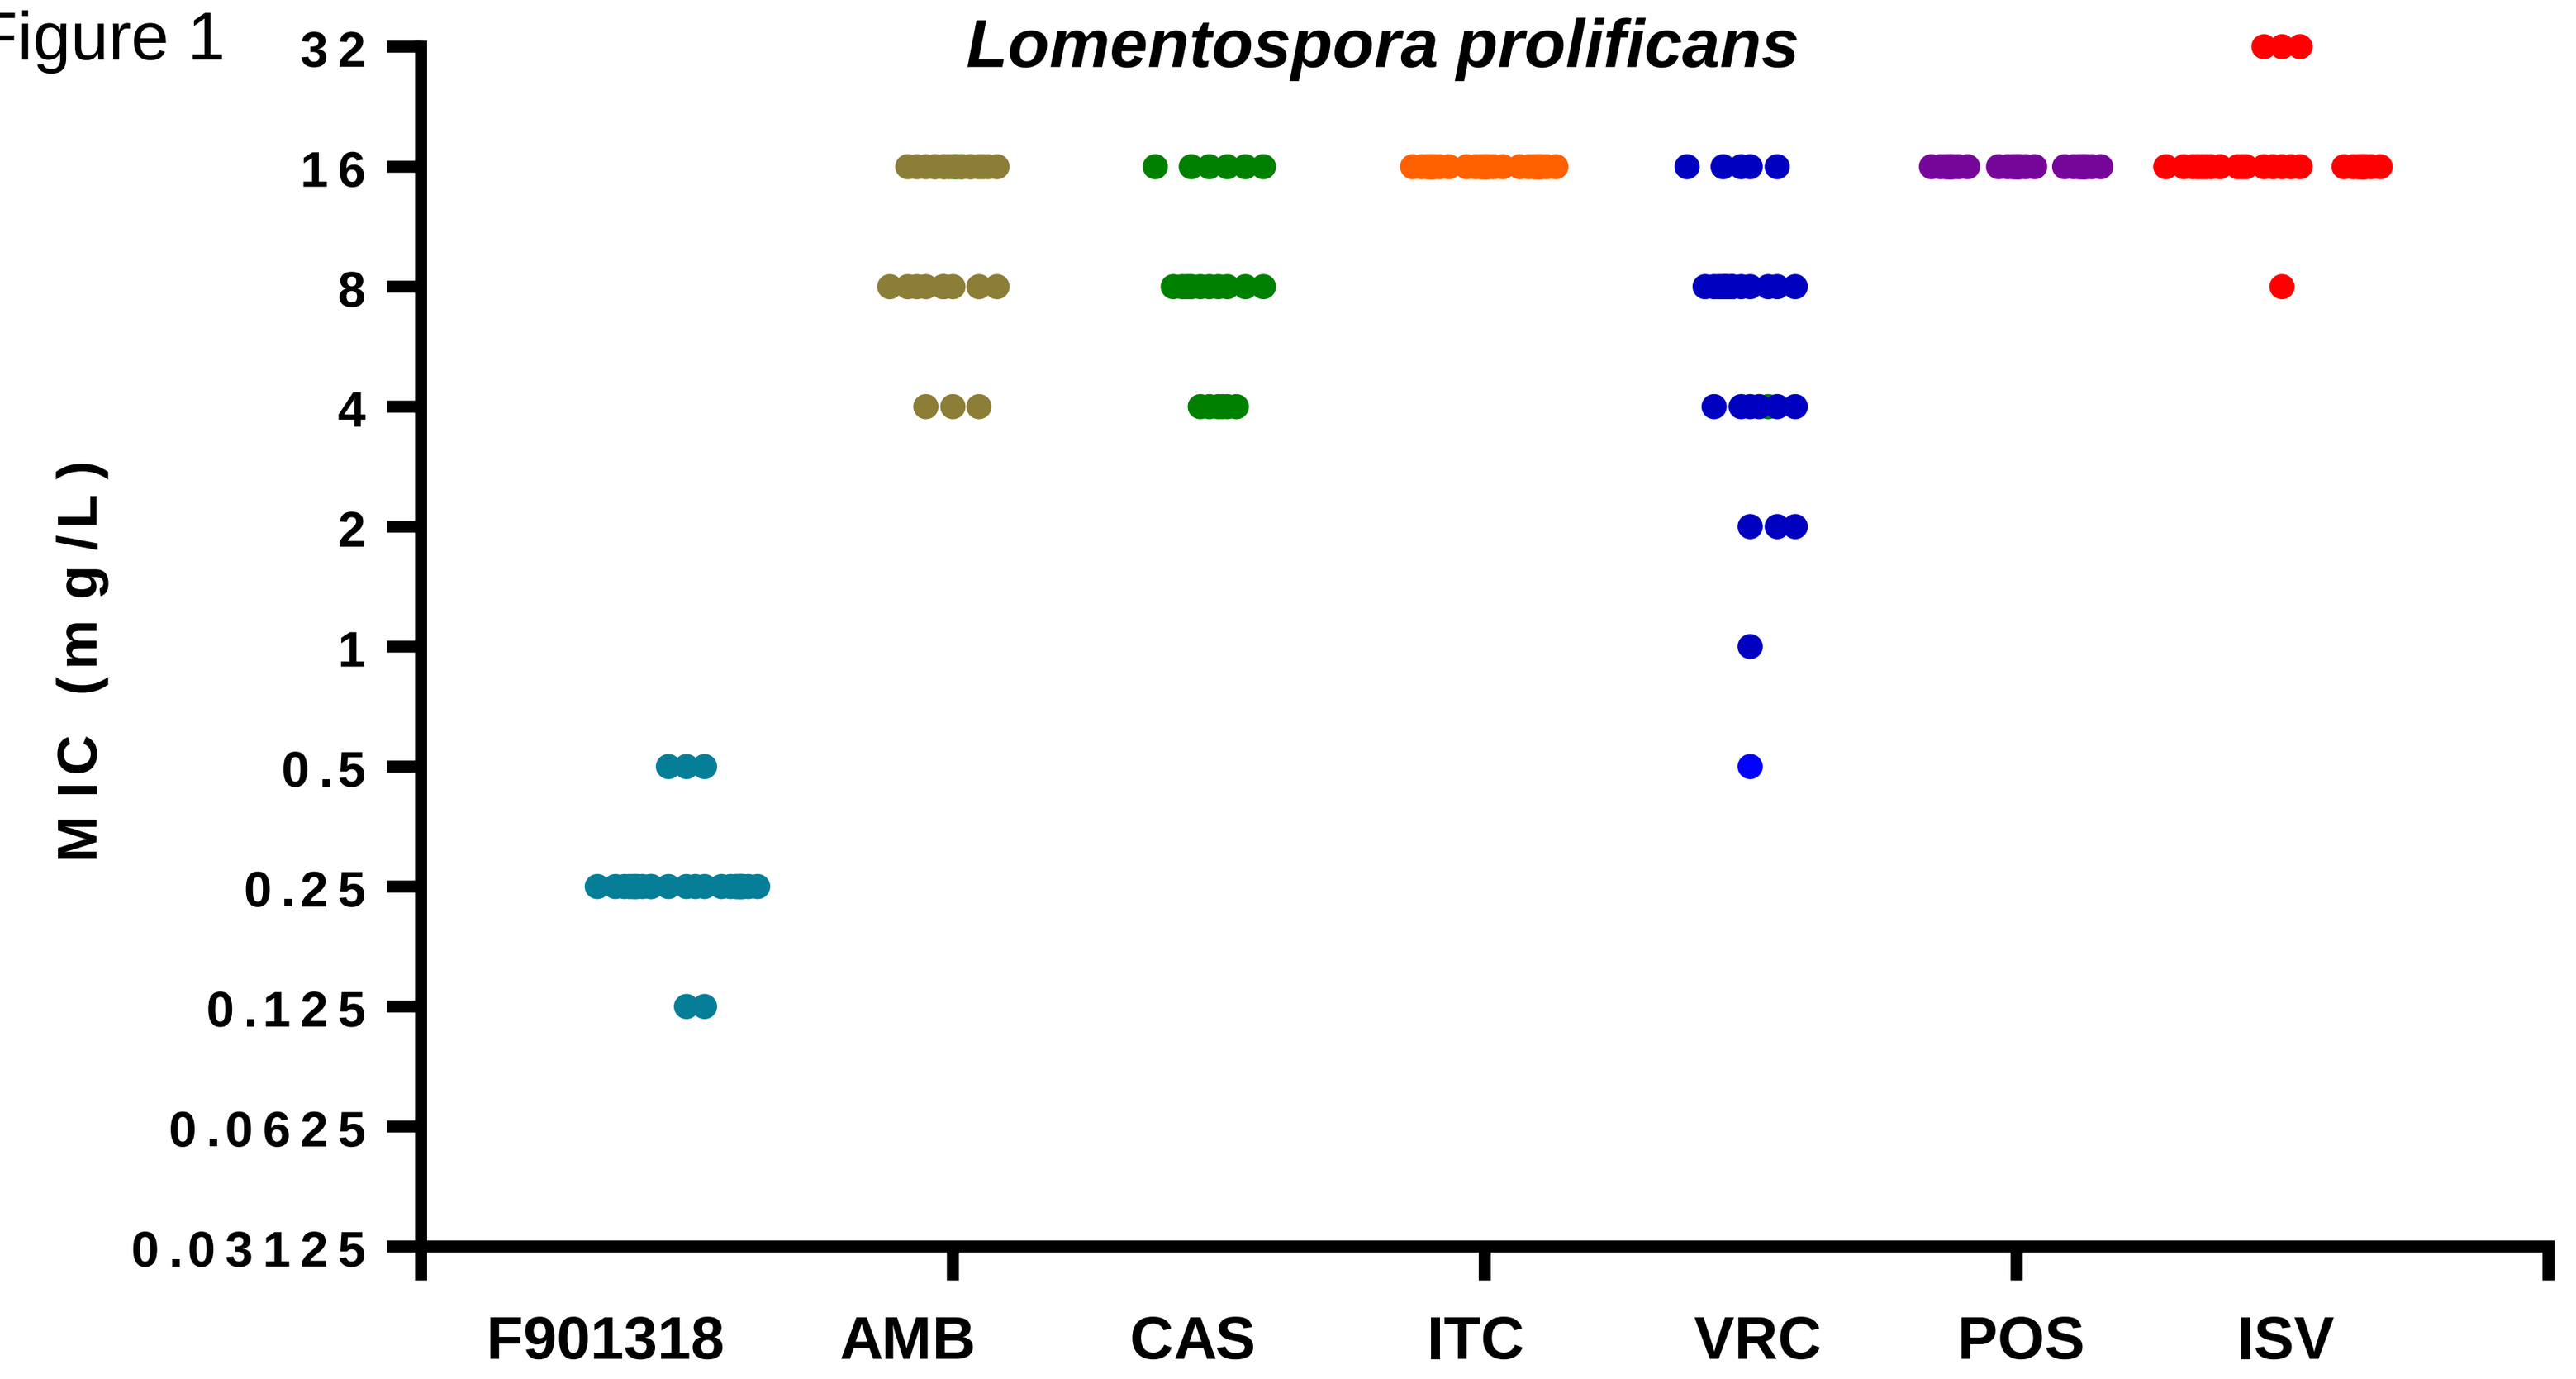
Lomentospora prolificans
MIC (mg/L)
32
16
8
4
2
1
0.5
0.25
0.125
0.0625
0.03125
F901318
AMB
CAS
ITC
VRC
POS
ISV

Figure 2

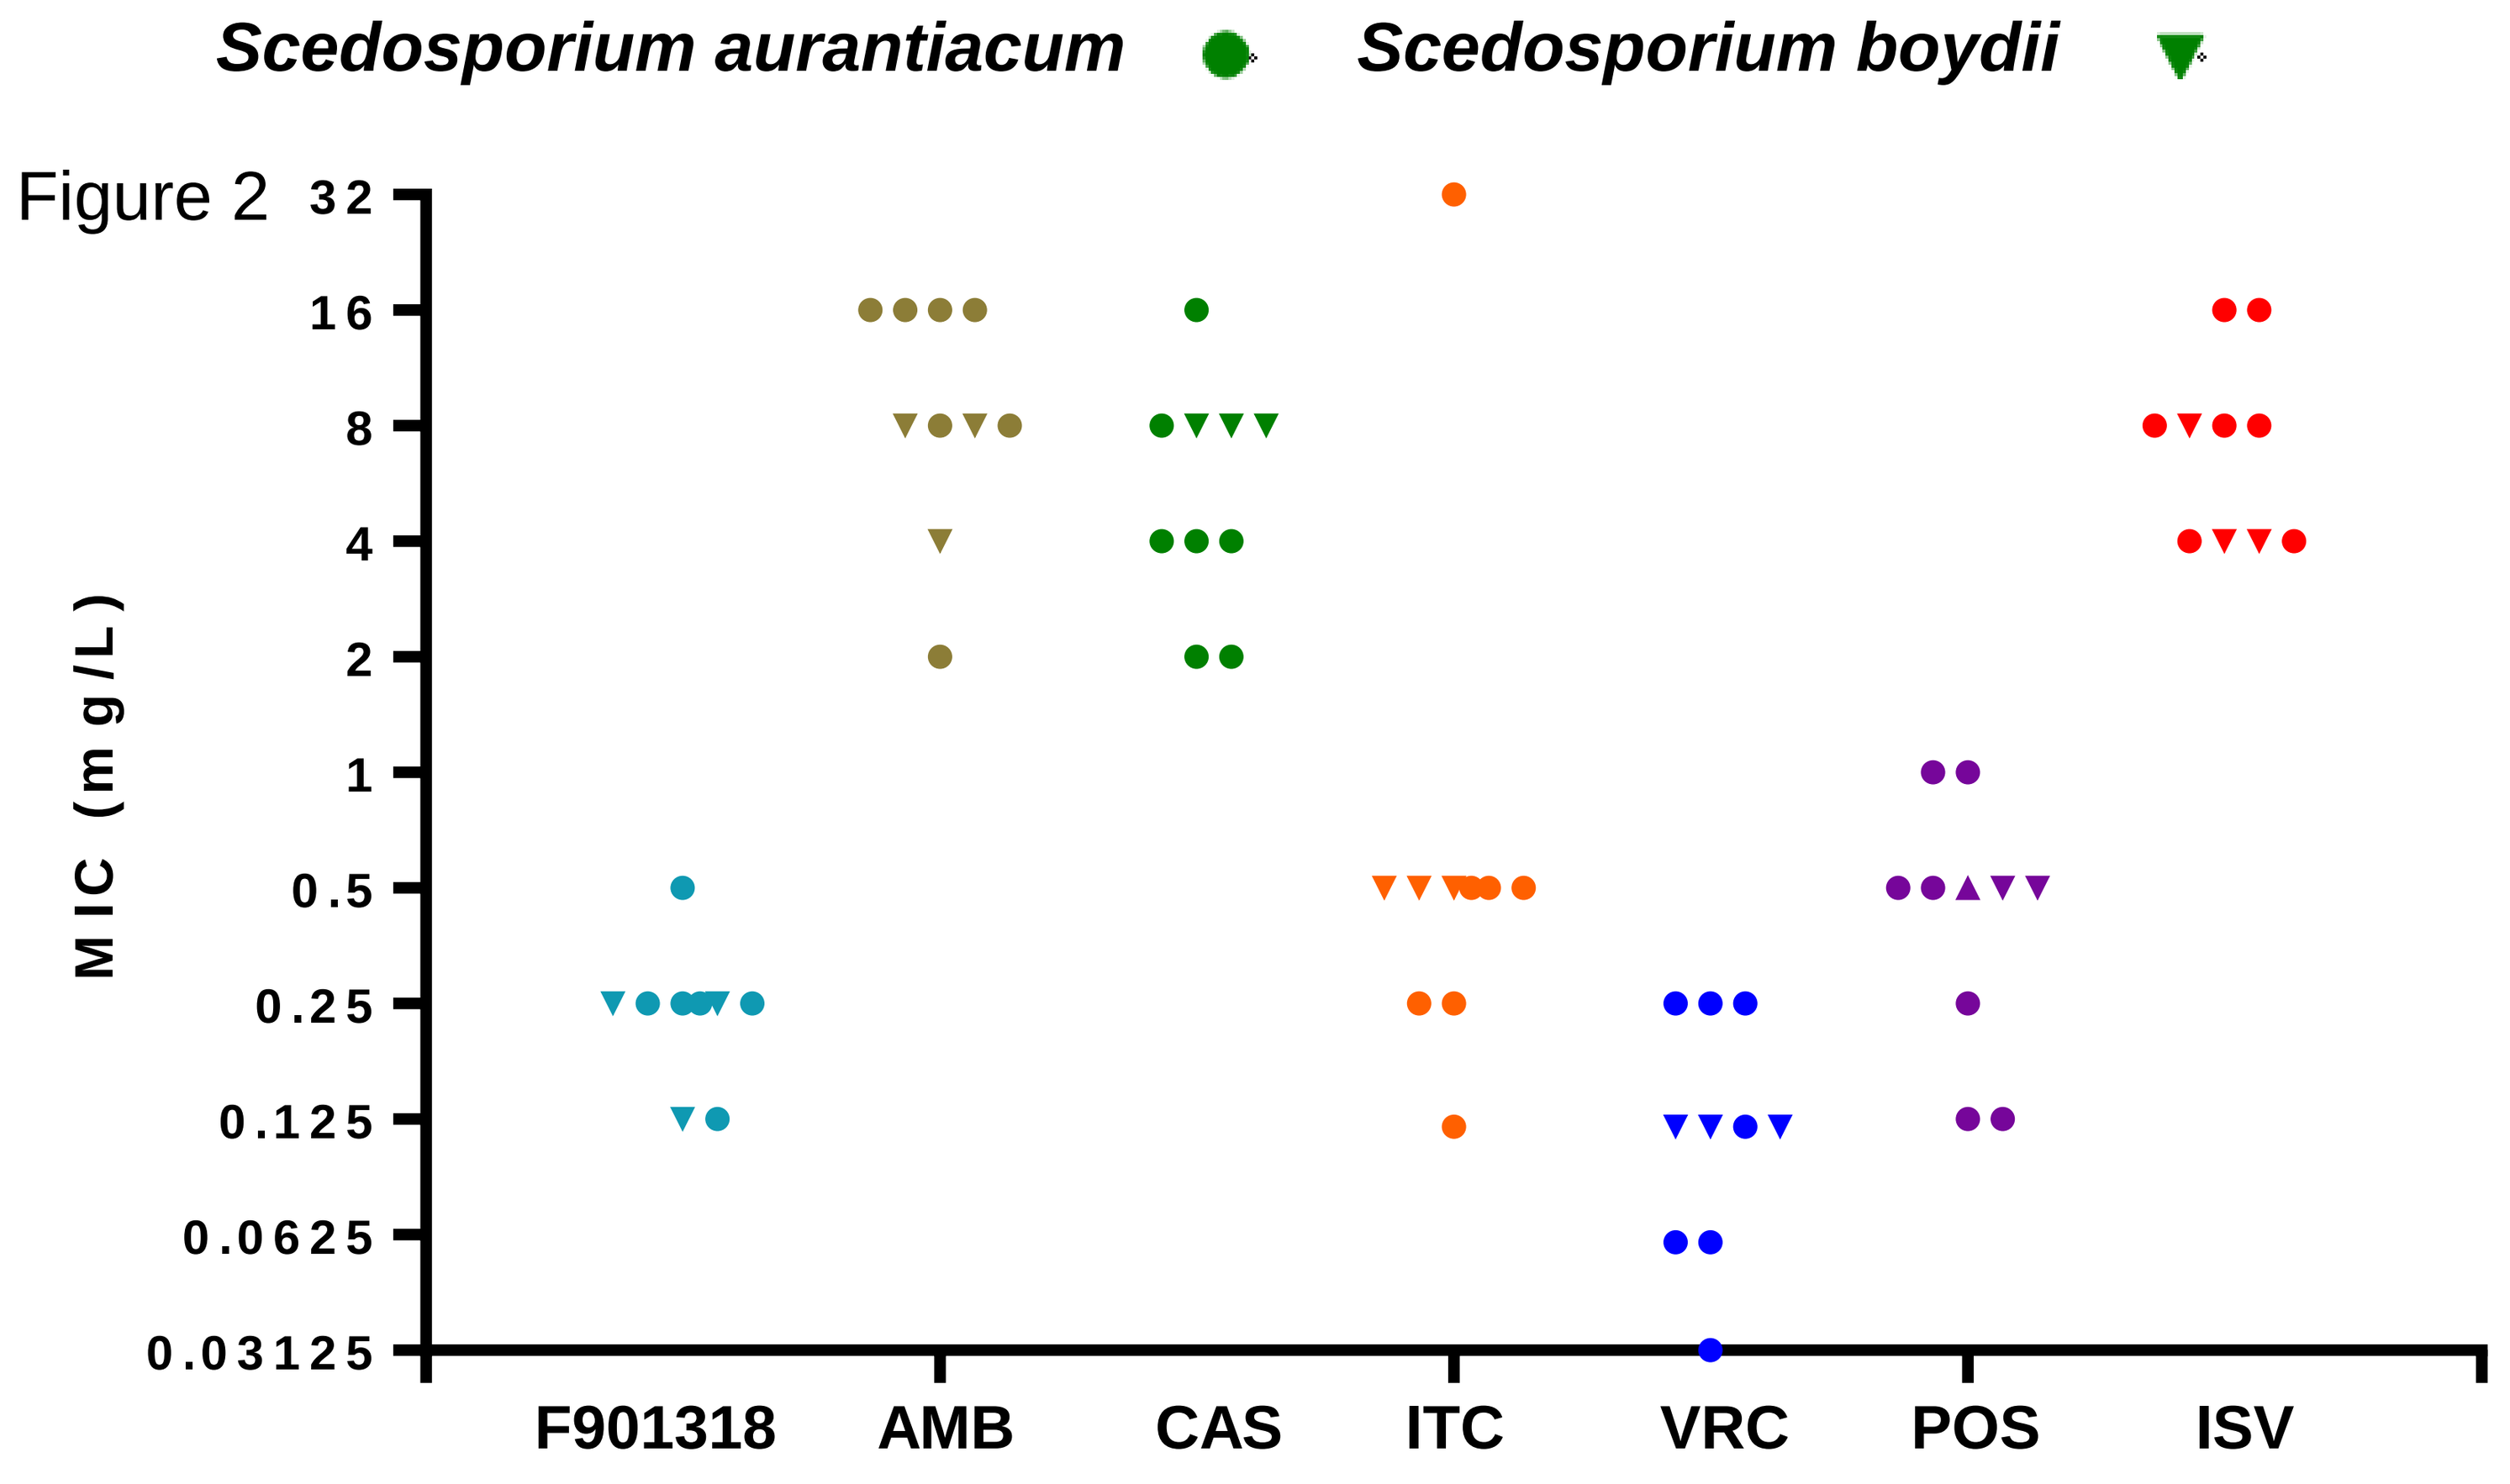
Scedosporium aurantiacum
Scedosporium boydii
MIC (mg/L)
32
16
8
4
2
1
0.5
0.25
0.125
0.0625
0.03125
F901318
AMB
CAS
ITC
VRC
POS
ISV

Figure 3

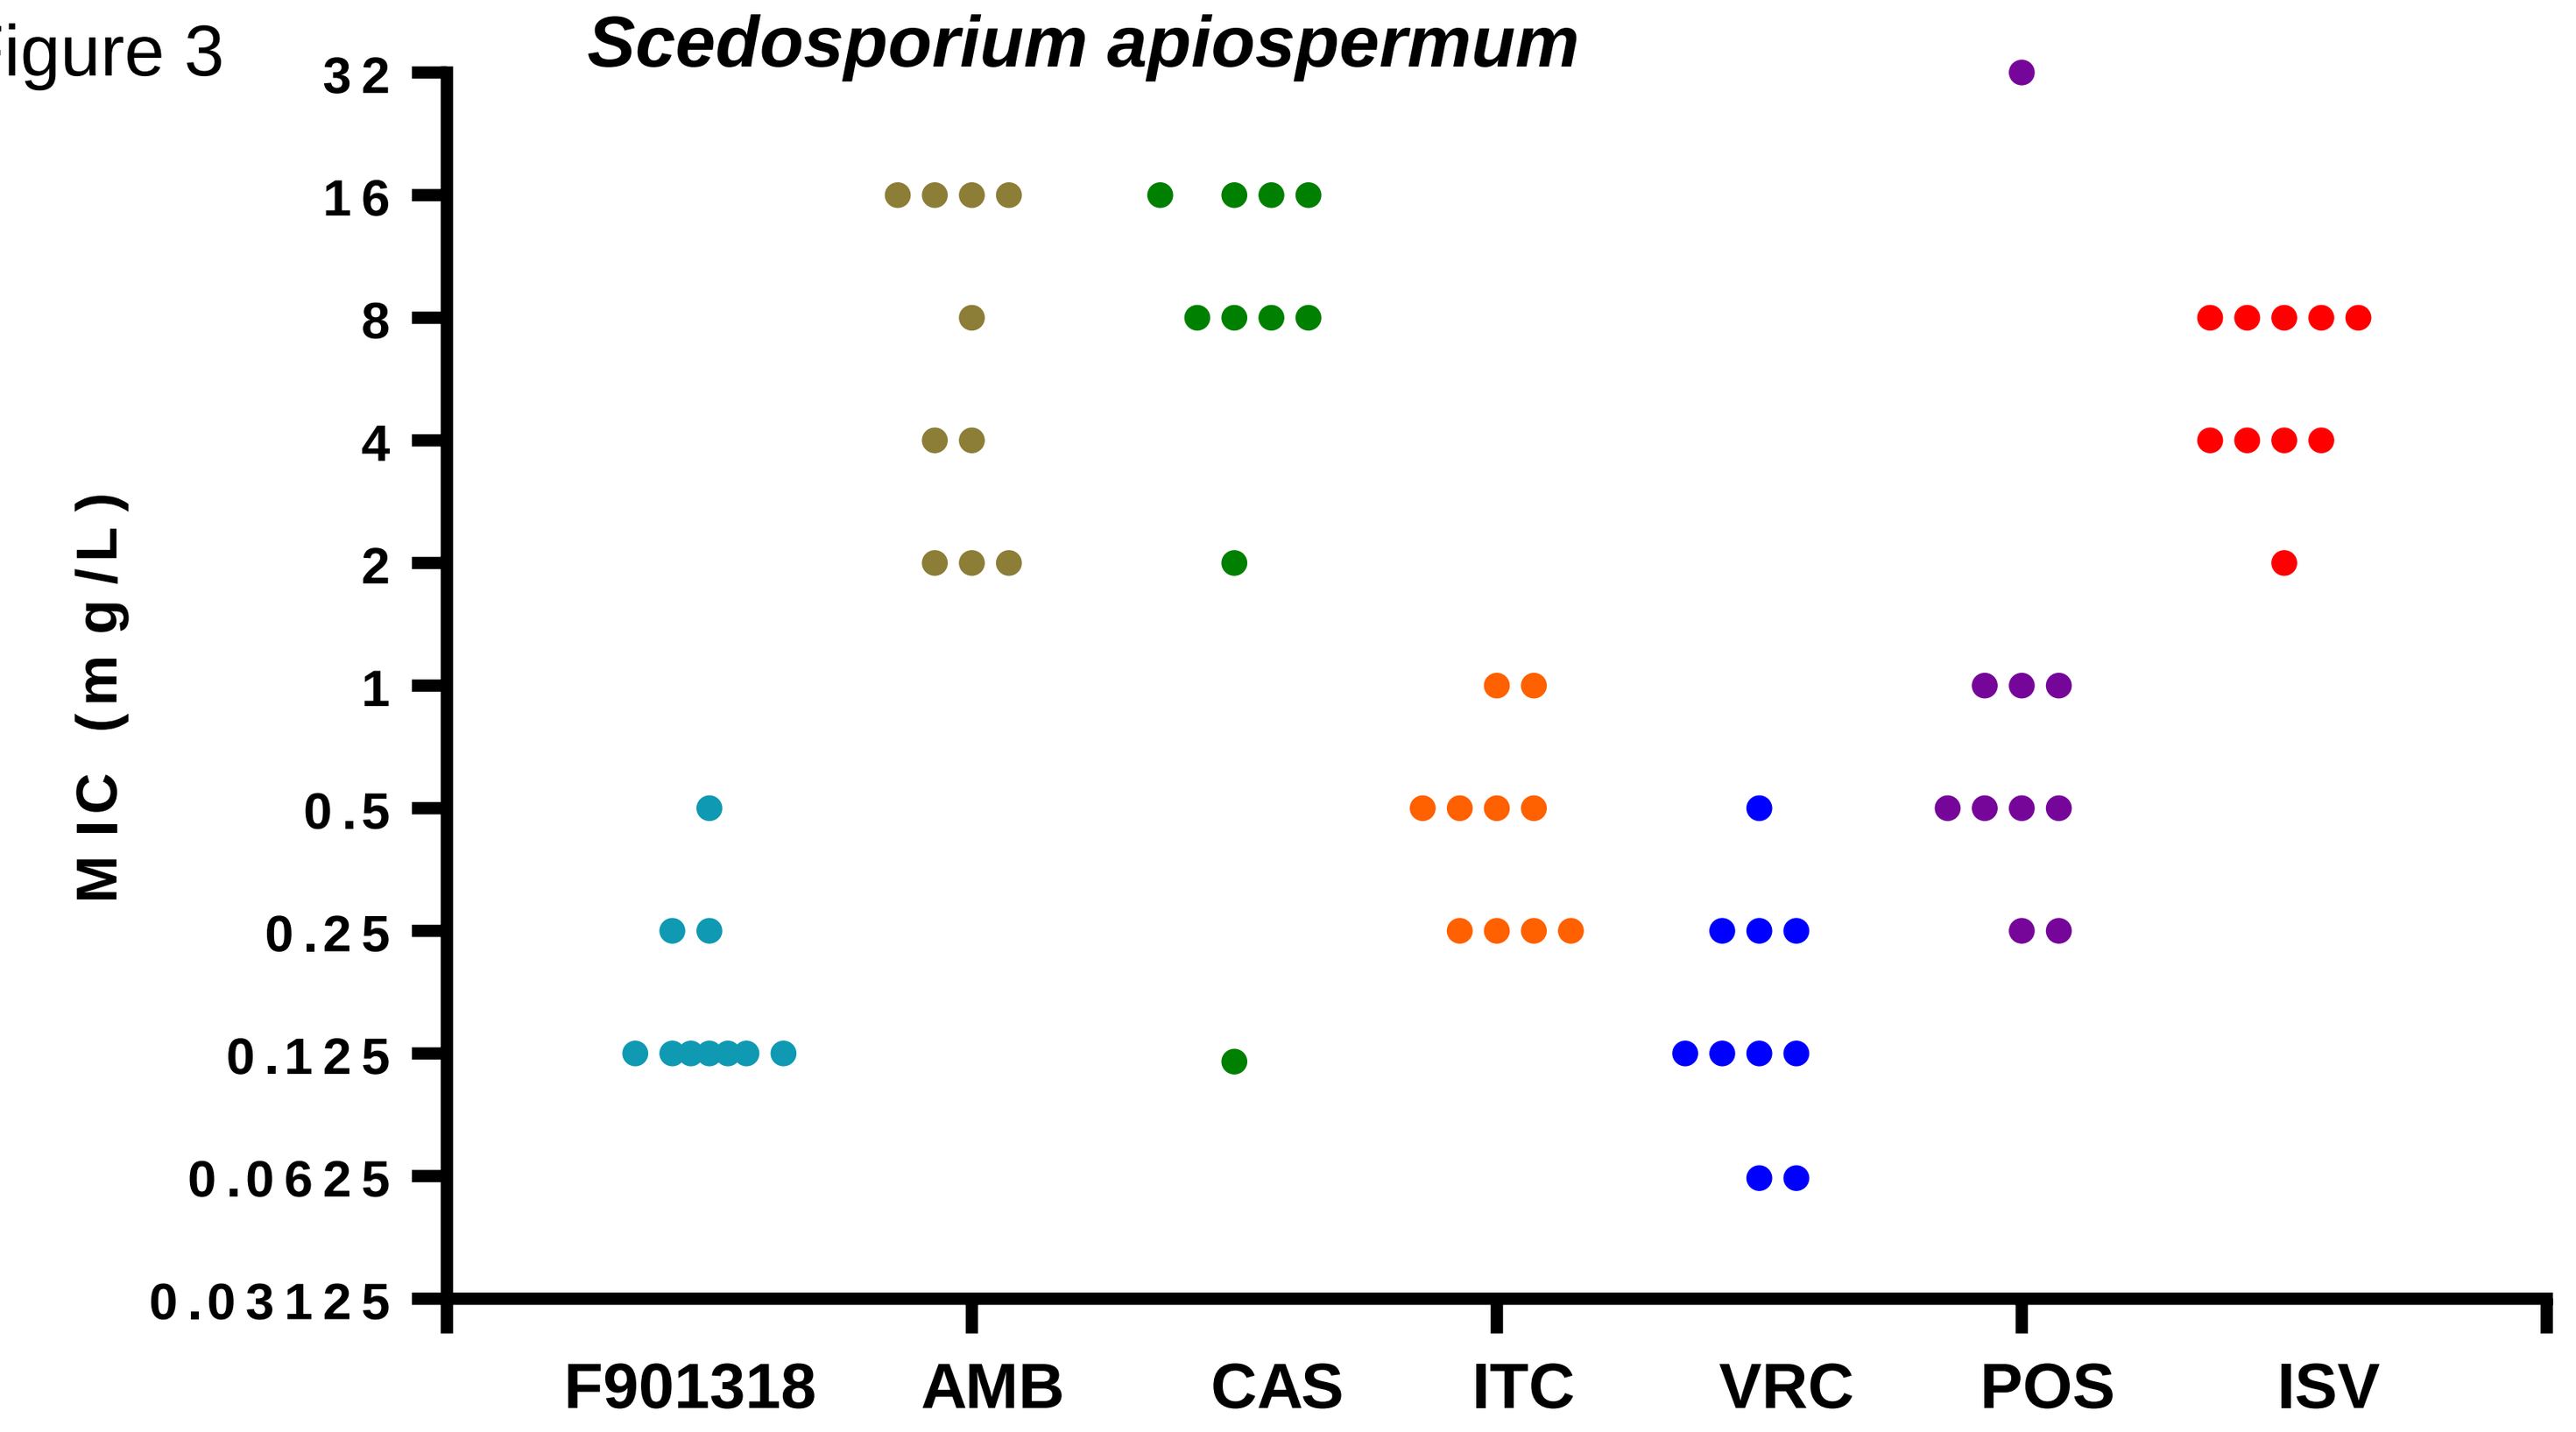
Scedosporium apiospermum
MIC (mg/L)
32
16
8
4
2
1
0.5
0.25
0.125
0.0625
0.03125
F901318
AMB
CAS
ITC
VRC
POS
ISV

Conclusion

1. F901318 demonstrated potent *in vitro* activity against 20 strains representing three species of *Scedosporium* and 30 strains of *L. prolificans*, and was the only agent to show consistently low MICs (0.125-0.5 mg/L) against the latter.

2. F901318 MICs for *Scedosporium* and *Lomentospora* spp. were very similar to those obtained for 4 *A. fumigatus* isolates, where animal models show potent F901318 *in vivo* activity.

3. The results for a relatively large panel of Australian *L. prolificans* clinical isolates highlight the poor activity of all approved antifungals, and underscore the need to further pursue the clinical applicability of F901318 as a novel antifungal agent for *Lomentospora*/*Scedosporium* spp. infections.