The Novel Orotomide F901318 Demonstrates Potent *In vitro* Antifungal Activity Against *Lomentospora* and *Scedosporium* Species

WE MAKE LIVES BETTER
UT HEALTH SCIENCE CENTER™
SAN ANTONIO

A. W. Fothergill,[1] D. Law,[2] M. Birch,[2] N.P. Wiederhold[1]

[1]UT Health Science Center San Antonio Fungus Testing Laboratory, [2]F2G Ltd., Manchester, United Kingdom

P-1575

Contact Information:
N.P. Wiederhold,
UTHSCSA MSC 7750
7703 Floyd Curl Dr.,
San Antonio, TX 78229
Tel: (210) 567-4086
e-mail: wiederholdn@uthscsa.edu

ABSTRACT

Background: *Scedosporium* species and *Lomentospora* (formerly *Scedosporium*) *prolificans* are increasing causes of invasive fungal infections in immunocompromised hosts. Many isolates of these species are resistant to available antifungals, and treatment options are limited. F901318 is the most advanced analog of the orotomide class of antifungal agents with a novel mechanism of action and is currently under clinical development. The objective of this study was to assess the *in vitro* potency of F901318 against *Scedosporium* species and *L. prolificans*.

Material/methods: Sixty-six *Scedosporium* isolates and 7 *Lomentospora prolificans* clinical isolates were used in this study. This included 43 *S. apiospermum,* 15 *S. boydii*, 6 *S. aurantiacum*, 2 *S. dehoogii*, and 7 *L. prolificans* isolates. The species identification of each isolate was confirmed by morphologic assessment and DNA sequence analysis of the ITS region and calmodulin gene. *In vitro* susceptibility testing was performed according to the methods in the CLSI M38-A2 reference standards. For F901318, amphotericin B, posaconazole, and voriconazole MICs were read after 72 hours of incubation as the lowest concentration that resulted in 100% inhibition of growth. For caspofungin the MEC was read as the lowest concentration that resulted in morphologic changes (i.e., short, stubby hyphae with abnormal branching). The MIC50, MIC90, and geometric mean (GM) MIC/MEC were determined, and differences in GM MIC values were assessed for significance by ANOVA.

Results: F901318 demonstrated the most potent *in vitro* activity of all the agents included in this study. Against *S. apiospermum* and *boydii* the F901318 GM MICs (0.079 and 0.046 µg/mL, repectively) were significantly lower than those observed with amphotericin (3.404 and 5.595 µg/mL , posaconazole (1.937 and 1.823 µg/mL), voriconazole (0.784 and 0.630 µg/mL), and caspofungin (5.703 and 7.639 µg/mL) (p < 0.001 for all comparisons). MIC50 and MIC90 values for F901318 were also lower for *S. apiospermum* and *S. boydii* than those of the comparator agents. Against *S. aurantiacum* and *S. dehoogii* isolates the F901318 MIC range (0.12 – 0.5 µg/mL) was also lower than the MIC/MEC ranges for the other antifungals (0.5 - >8 µg/mL). The activity of F901318 also maintained against *L. proflicans* isolates (range 0.12 – 0.25 µg/mL) in contrast to that observed with the other antifungals, none of which demonstrated *in vitro* activity against this multidrug resistant species.

Conclusions: F901318, a novel member of the orotomide class of antifungals, demonstrated potent *in vitro* activity against *Scedosporium* species and *L. prolificans*. This activity was maintained against isolates that had significantly reduced susceptibility to the other antifungals included in this study, including *L. prolificans,* for which treatment options are limited. Further studies are warranted to evaluate the in vivo efficacy of F901318 against *Scedosporium* species and *L. prolificans*.

BACKGROUND

- *Scedosporium* species and *Lomentospora* (formerly *Scedosporium*) *prolificans* are increasing causes of invasive fungal infections in immunocompromised hosts.
- Many isolates of these species are resistant to available antifungals, and treatment options are limited.
- F901318 is the most advanced analog of the orotomide class of antifungal agents and is currently under clinical development.
- This investigational agent acts via a novel mode of action namely the selective inhibition of DHODH a critical enzyme in pyrimidine biosynthesis.

OBJECTIVE

The objective of this study was to assess the *in vitro* potency of F901318 against *Scedosporium* species and *L. prolificans*. The *in vitro* activity of F901318 was compared to that of the clinically available antifungals amphotericin B, caspofungin, voriconazole, and posaconazole.

MATERIALS AND METHODS

Fungal Isolates

•Clinical isolates of *Scedosporium* spp. and *Lomentosporum prolificans* that were submitted to the Fungus Testing Laboratory at the UT Health Science Center at San Antonio were used. These consisted of 43 *S. apiospermum*, 6 *S. aurantiacum,* 15 *S. boydii*, 2 *S. dehoogii*, and 7 *L. prolificans.*

•All isolates were identified to the species level by combined morphologic/phenotypic assessment and DNA sequence analysis of the following targets: ITS and calmodulin.

Antifungal Susceptibility Testing

•Antifungal susceptibility testing was performed by broth microdilution in RPMI buffered with 0.165M MOPS (pH 7.0) according to the CLSI M38-A2 reference method.

•Antifungal agents included F901318, amphotericin B (AMB), caspofungin (CAS), posaconazole (POS), and voriconazole (VOR). Stocks of each agent were prepared in DMSO (final DMSO concentration 1% in microtiter trays), and the concentration ranges were 0.008 – 4 µg/mL for F901318, 0.015 – 8 µg/mL for caspofungin, and 0.03 – 16 µg/mL for amphotericin B, posaconazole, and voriconazole.

•Minimum inhibitory concentrations (MICs) for F901318, amphotericin B, posaconazole, and voriconazole were read after 72 hours of incubation at 35ºC as the lowest concentration that resulted in 100% inhibition of growth. The minimum effective concentration (MEC) for caspofungin was read as the lowest concentration that resulted in a morphological change compared to growth control (i.e., abnormally branched, short, stubby hyphae).

Data Analysis

•The MIC and MEC ranges, MIC and MEC values that inhibited 50% and 90% of the isolates (MIC50 and MIC90 values, respectively), and the geometric mean (GM) MICs were determined.

•MIC/MEC values greater than the highest concentration tested were assigned a value one dilution higher for graphing and statistical analysis.

•Differences in GM MIC values were assess for statistical significance by ANOVA with Tukey's post-test for multiple comparisons. A p-value < 0.05 was considered statistically significant.

RESULTS

Figure 1. Overall *in vitro* activity (µg/mL) of F901318, amphotericin B, caspofungin, posaconazole, and voriconazole against *Scedosporium* spp. (n = 66 clinical isolates).

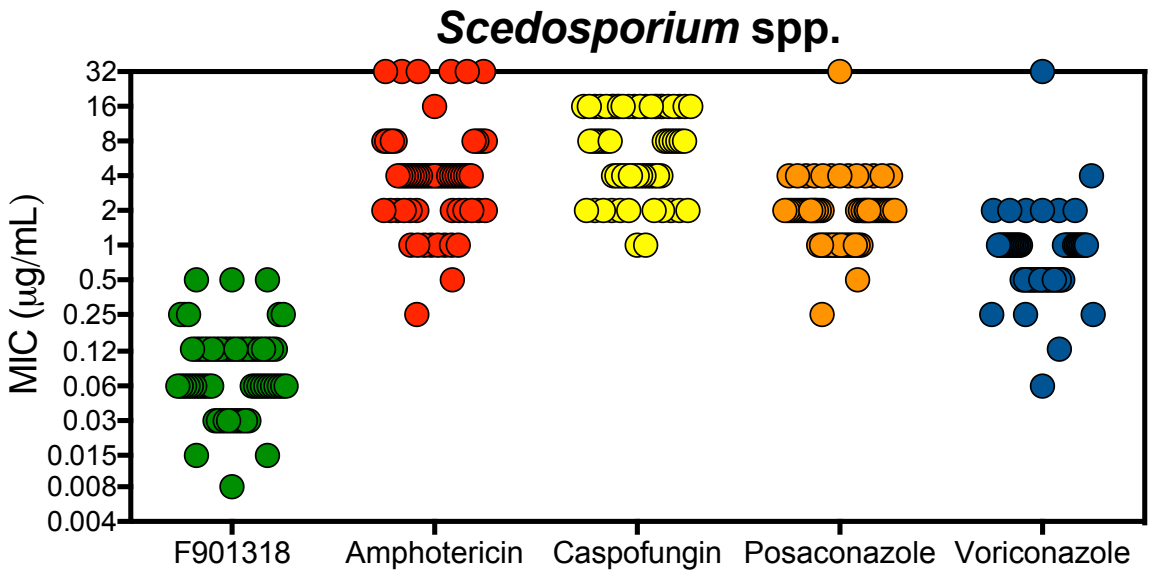

Scedosporium spp.
MIC (µg/mL)
32
16
8
4
2
1
0.5
0.25
0.12
0.06
0.03
0.015
0.008
0.004
F901318
Amphotericin
Caspofungin
Posaconazole
Voriconazole

Table 1. MIC ranges, MIC50 & MIC90 values, and GM MICs for F901318, amphotericin B, caspofungin, posaconazole, and voriconazole against *Scedosporium* spp. (n = 66 clinical isolates).

Antifungal	F901318	Amphotericin	Caspofungin	Posaconazole	Voriconazole
MIC Range	< 0.008 – 0.5	0.25 - >16	1 - >8	0.25 - >16	0.06 ->16
MIC50	0.06	4	8	2	1
MIC90	0.25	16	>8	4	2
GM MIC	0.078	3.795*	6.553*	1.918	0.769

*P-value < 0.05 vs. F901318

RESULTS (cont.)

Figure 2. *in vitro* activity (µg/mL) of F901318, amphotericin B, caspofungin, posaconazole, and voriconazole against clinical isolates of *S. apiospermum* (n = 43), *S. aurantiacum* & *S. dehoogii* (n = 8), *S. boydii* (n = 15) and *L. prolificans* (n = 7). *S. dehoogii* MICs shown in gray circles.

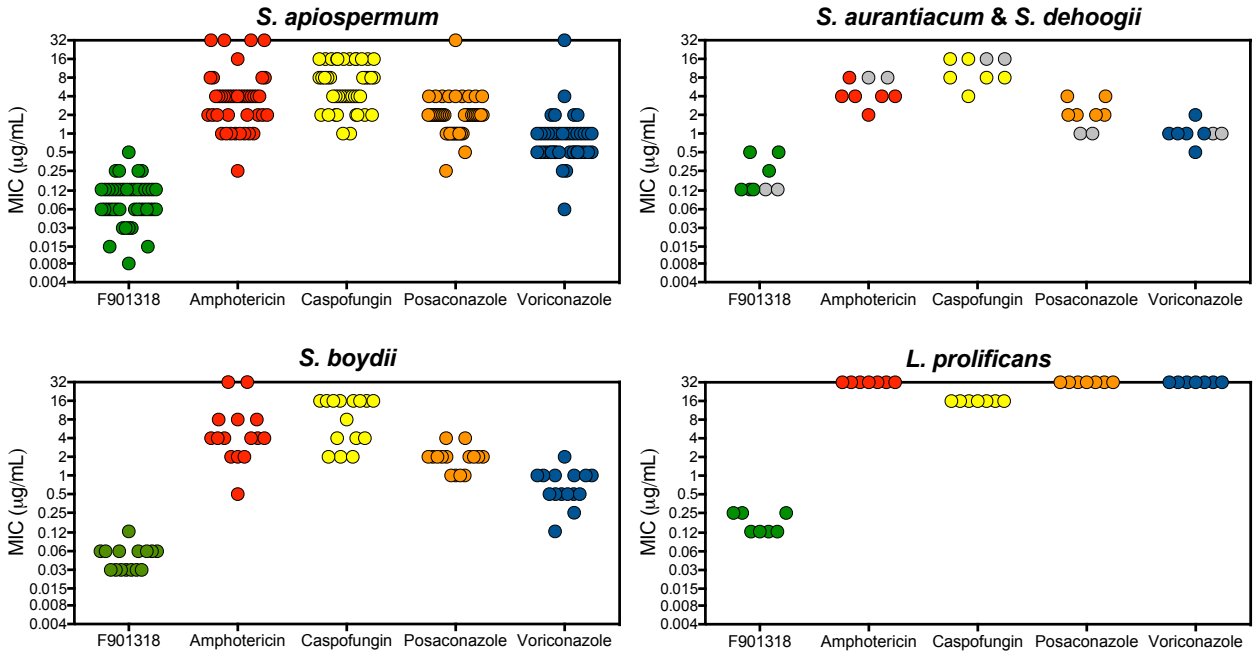

S. apiospermum
S. aurantiacum & S. dehoogii
S. boydii
L. prolificans
MIC (µg/mL)
F901318
Amphotericin
Caspofungin
Posaconazole
Voriconazole

Table 1. MIC ranges, MIC50 & MIC90 values, and GM MICs for F901318, amphotericin B, caspofungin, posaconazole, and voriconazole against clinical isolates of *S. apiospermum* (n = 43), *S. aurantiacum* & *S. dehoogii* (n = 8), *S. boydii* (n = 15) and *L. prolificans* (n = 7).

Antifungal	F901318	Amphotericin	Caspofungin	Posaconazole	Voriconazole
Scedosporium apiospermum (n = 43)					
MIC Range	< 0.008 – 0.5	0.25 - >16	1 - >8	0.25 - >16	0.06 ->16
MIC50	0.06	4	8	2	1
MIC90	0.25	16	>8	4	2
GM MIC	0.079	3.404*	5.703*	1.937	0.784
Scedosporium aurantiacum (n = 6) & *S. dehoogii* (n = 2)					
MIC Range	0.12 – 0.25	2 - 8	4 - >8	1 - 4	0.5 - 2
MIC50	0.12	4	8	2	1
MIC90	0.5	8	>8	4	2
GM MIC	0.193	4.757*	>8*	2.000	1.000
Scedosporium boydii (n = 15)					
MIC Range	< 0.008 – 0.12	0.5 - >16	2 - >8	1 - 4	0.12 - 2
MIC50	0.06	4	>8	2	0.5
MIC90	0.06	>16	>8	4	1
GM MIC	0.046	5.595*	7.639*	1.832	0.630
Lomentospora prolificans (n = 7)					
MIC Range	0.12 – 0.25	>16	>8	>16	>16
MIC50	0.12	>16	>8	>16	>16
MIC90	0.25	>16	>8	>16	>16
GM MIC	0.168	>16*	>8*	>16*	>16*

*P-value < 0.05 vs. F901318

CONCLUSIONS

F901318, a novel member of the orotomide class of antifungals, demonstrated potent *in vitro* activity against *Scedosporium* species and *L. prolificans*. This activity was maintained against isolates that had significantly reduced susceptibility to the other antifungals used in this study, including *L. prolificans,* for which treatment options are limited. Further studies are warranted to evaluate the *in vivo* efficacy of F901318 against *Scedosporium* species and *L. prolificans.*

Funding and F901318 powder was provided by F2G, Ltd.